P1123

Matching-Adjusted Indirect Comparison of Zanubrutinib Versus Real-World Chemoimmunotherapy or Chemotherapy in Relapsed/Refractory Marginal Zone Lymphoma

Renata Walewska,[1] Kaijun Wang,[2] Caitlin Smare,[3] Ina Zhang,[4] Leyla Mohseninejad,[5] Silvy Mardiguian,[6] Sam Keeping[7]

[1]University Hospitals Dorset, Department of Haematology, Bournemouth, UK; [2]BeiGene USA, Inc, San Mateo, CA, USA; [3]PRECISIONheor, London, UK; [4]PRECISIONheor, Oakland, CA, USA; [5]BeiGene Netherlands B.V., Schiphol, The Netherlands; [6]BeiGene, London, UK; [7]PRECISIONheor, Vancouver, British Columbia, Canada

INTRODUCTION

- Marginal zone lymphoma (MZL), a form of non-Hodgkin lymphoma (NHL), can be difficult to treat due to its rarity and range of clinical presentation
- Patients with advanced disease are characterized by a continuing pattern of relapse and remission; current management for these patients generally involves utilizing regimens shown to be effective in other indolent NHLs such as repeating or alternating chemoimmunotherapy (CIT), immunotherapy alone, or chemotherapy (chemo) regimens
- Zanubrutinib, a next-generation Bruton tyrosine kinase inhibitor, has recently been approved for the treatment of relapsed/refractory (R/R) MZL in the European Union (EU) and United States (US) based on two phase 2 single-arm trials, MAGNOLIA (NCT03846427) and BGB-3111-AU-003 (NCT02343120)
- Two previous matching-adjusted indirect comparisons (MAICs) have shown zanubrutinib to be more efficacious in terms of progression-free survival (PFS) vs both ibrutinib (PCYC-1121) and rituximab (CHRONOS-3) based on data from clinical trials[1,2]
- In the absence of head-to-head randomized controlled trials comparing zanubrutinib vs other treatment choices, unanchored indirect treatment comparisons can be utilized to estimate relative treatment effects

OBJECTIVE

- This study aimed to estimate the comparative efficacy of zanubrutinib vs CIT, immunotherapy, or chemo for the treatment of patients with R/R MZL by conducting a MAIC with data from the single arm trials of zanubrutinib and real-world data of CIT, immunotherapy, or chemo use

METHODS

Data Sources

- Individual patient-level data from 86 efficacy-evaluable patients enrolled in MAGNOLIA and BGB-3111-AU-003 trials were used to inform the zanubrutinib treatment group[3,4]
- Aggregate data from a comparable cohort of 90 patients treated with CIT, immunotherapy, or chemo was identified from the Haematological Malignancy Research Network (HMRN), a UK cancer registry[5]
- All patients in the HMRN cohort were enrolled from 2014 onward, had an Eastern Cooperative Oncology Group Performance Status (ECOG PS) ≤2 at entry into the registry, and were previously treated with an anti-CD20-based therapy, in line with the inclusion and exclusion criteria of MAGNOLIA and BGB-3111-AU-003 trials
- All patients in the HMRN cohort were treated with CIT, immunotherapy, or chemo alone; the most commonly used treatments being bendamustine + rituximab (30%), rituximab monotherapy (13%), and rituximab, cyclophosphamide, vincristine, and prednisolone (11%)

Statistical Analysis

- Logistic propensity score models were used to estimate weights for the pooled population of patients from MAGNOLIA and BGB-3111-AU-003 such that mean baseline characteristics matched those in the HMRN cohort
- Prespecified characteristics for matching were confirmed by clinical experts and are presented in order of importance in **Table 1**
- As patients receiving chemo alone were anticipated to have worse outcomes than those receiving CIT or immunotherapy, a sensitivity analysis was performed whereby 17 patients treated with chemo only were excluded from the model
- Relevant outcomes were PFS and overall survival (OS); PFS was assessed by an independent review committee in the zanubrutinib trials and disease progression data in the HMRN cohort was collected from medical records
- Cox proportional hazards models were used to estimate relative treatment effects in terms of hazard ratios (HRs) and their 95% confidence intervals (CIs)
- Leave-one-out analyses were performed, whereby one covariate at a time was omitted from the model to explore their individual impact on the treatment effect estimates

RESULTS

- Before matching, the zanubrutinib population consisted of slightly younger, more heavily pretreated patients, with a higher proportion who were refractory and who on average had a longer time since diagnosis than the HMRN cohort
- Convergence was achieved for all the logistic propensity score models resulting in baseline characteristics that were balanced between the treatment groups after matching (**Table 1**); the effective sample size (ESS) was 38 (a 56% reduction from a sample of 86)
- Results from the MAIC (with unadjusted comparisons also presented for informative purposes only) are reported in **Table 2**

Table 1. Comparison of Patient Characteristics Before and After Matching

Characteristic		Zanubrutinib (MAGNOLIA & BGB-3111-AU-003 pooled)		CIT, immunotherapy, or chemo (HMRN)
		Unweighted, n=86	Weighted, ESS=38	n=90
Number of prior therapies, %	1	44.2	78.9	78.9
	2	30.2	18.9	18.9
	>2	25.6	2.2	2.2
Refractory to last therapy, %		30.1	25.6	25.6
POD24, %		44.7	51.1	51.1
Mean age, years		68.0	73.3	73.3
Time since diagnosis ≥ median, %		65.1	50.0	50.0

POD24, progression of disease within 24 months of treatment initiation.

Table 2. Results of Indirect Treatment Comparisons

Model	Zanubrutinib n/ESS	PFS HR (95% CI) *P* value	OS HR (95% CI) *P* value
Zanubrutinib vs CIT, immunotherapy, or chemo (n=90)			
Unadjusted	86	0.47 (0.29-0.76) *P*<.01	0.34 (0.19-0.61) *P*<.01
MAIC (all covariates)	38	0.30 (0.15-0.63) *P*<.01	0.23 (0.10-0.50) *P*<.01
MAIC (excluding number of prior lines)	49	0.48 (0.26-0.87) *P*=.02	0.37 (0.19-0.74) *P*<.01
MAIC (excluding refractory to last therapy)	39	0.29 (0.15-0.57) *P*<.01	0.22 (0.10-0.46) *P*<.01
MAIC (excluding age)	52	0.31 (0.16-0.59) *P*<.01	0.20 (0.09-0.43) *P*<.0001
MAIC (excluding POD24)	39	0.29 (0.14-0.62) *P*<.01	0.21 (0.09-0.45) *P*<.01
MAIC (excluding time since diagnosis)	41	0.26 (0.14-0.51) *P*<.01	0.24 (0.11-0.53) *P*<.01
Zanubrutinib vs CIT or immunotherapy (n=73)			
Unadjusted	86	0.51 (0.31-0.83) *P*<.01	0.37 (0.20-0.68) *P*<.01
MAIC (all covariates)	40	0.28 (0.14-0.57) *P*<.01	0.23 (0.10-0.49) *P*<.01

- Zanubrutinib significantly reduced the risk of progression (**Figure 1**) and death (**Figure 2**) relative to CIT, immunotherapy, and chemo. Model results excluding patients treated with chemo alone (ESS=40; 53% reduction) were consistent with these findings
- The leave-one-out analyses showed that removing any one of the characteristics did not significantly alter the treatment effect estimates, though number of prior lines of therapy had the largest impact

Figure 1. MAIC of PFS with Zanubrutinib vs CIT, immunotherapy, or Chemo

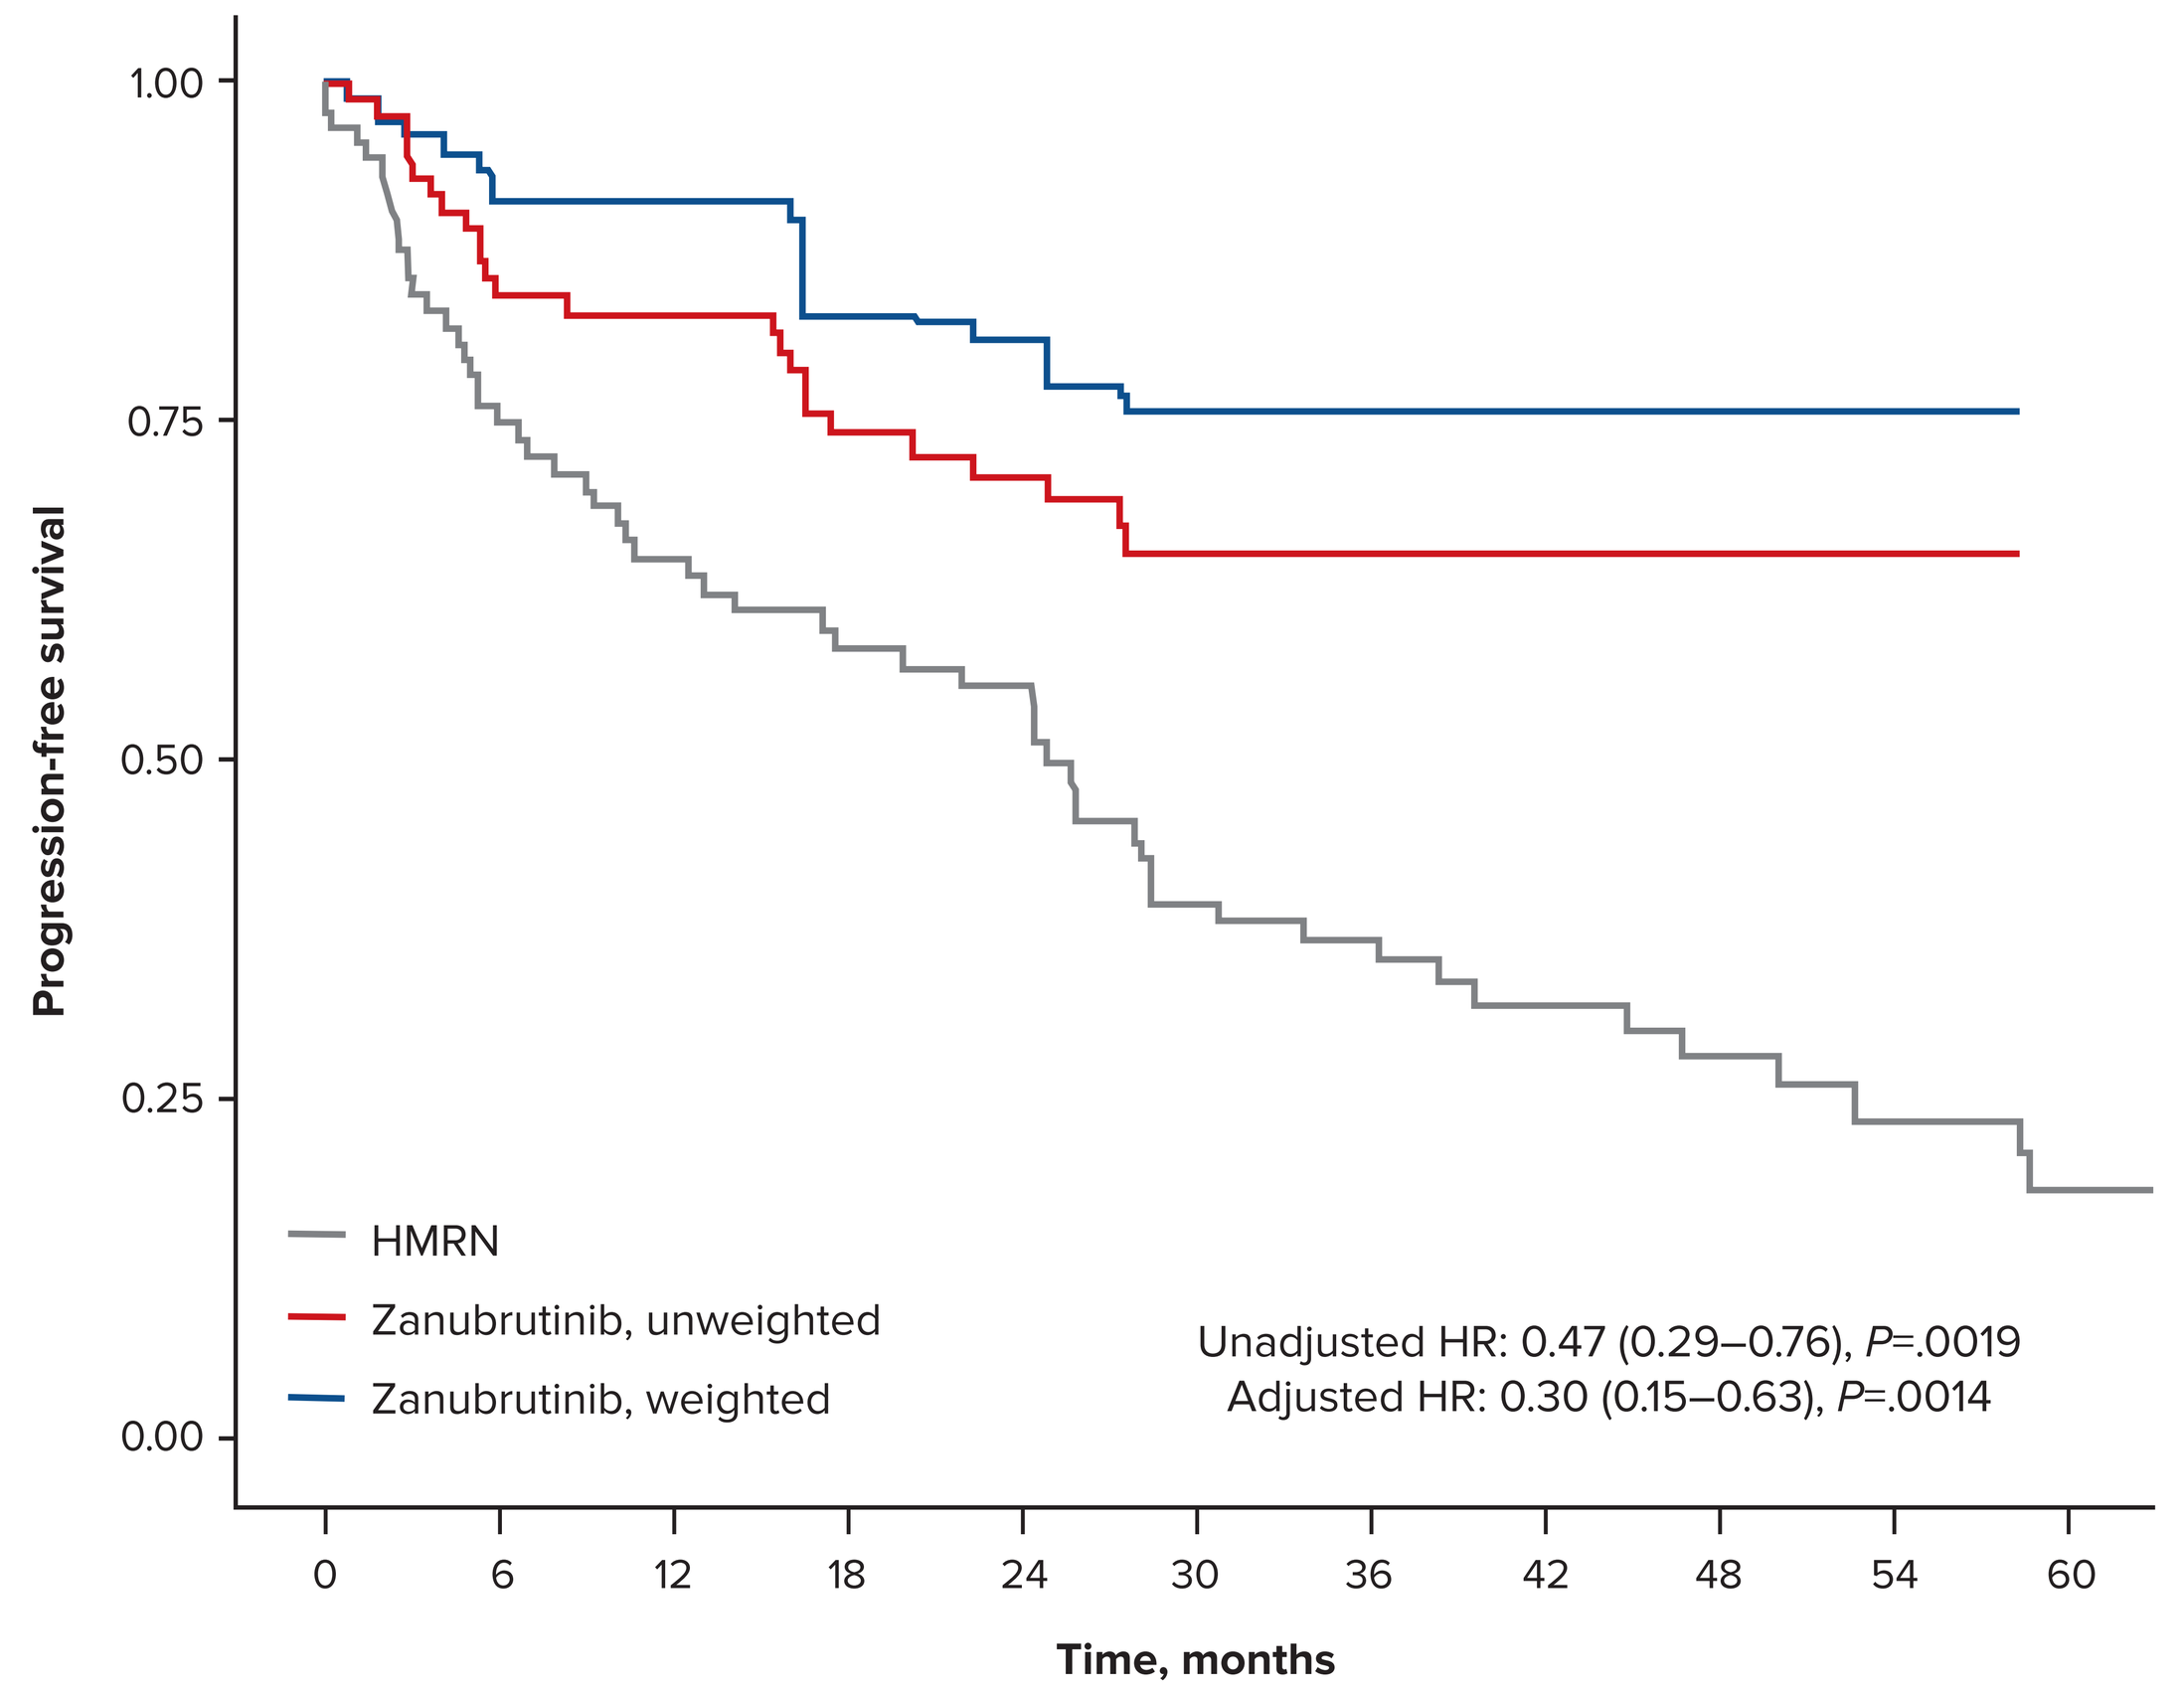
Progression-free survival
1.00
0.75
0.50
0.25
0.00
HMRN
Zanubrutinib, unweighted
Zanubrutinib, weighted
Unadjusted HR: 0.47 (0.29–0.76), P=.0019
Adjusted HR: 0.30 (0.15–0.63), P=.0014
0 6 12 18 24 30 36 42 48 54 60
Time, months

Number at risk

90	63	51	43	40	27	24	17	12	10	7
86	65	59	50	45	9	4	2	2	2	0
38	30	30	24	20	4	2	1	1	1	0

Values in parentheses are 95% CIs.

Figure 2. MAIC of OS with Zanubrutinib vs CIT, immunotherapy, or Chemo

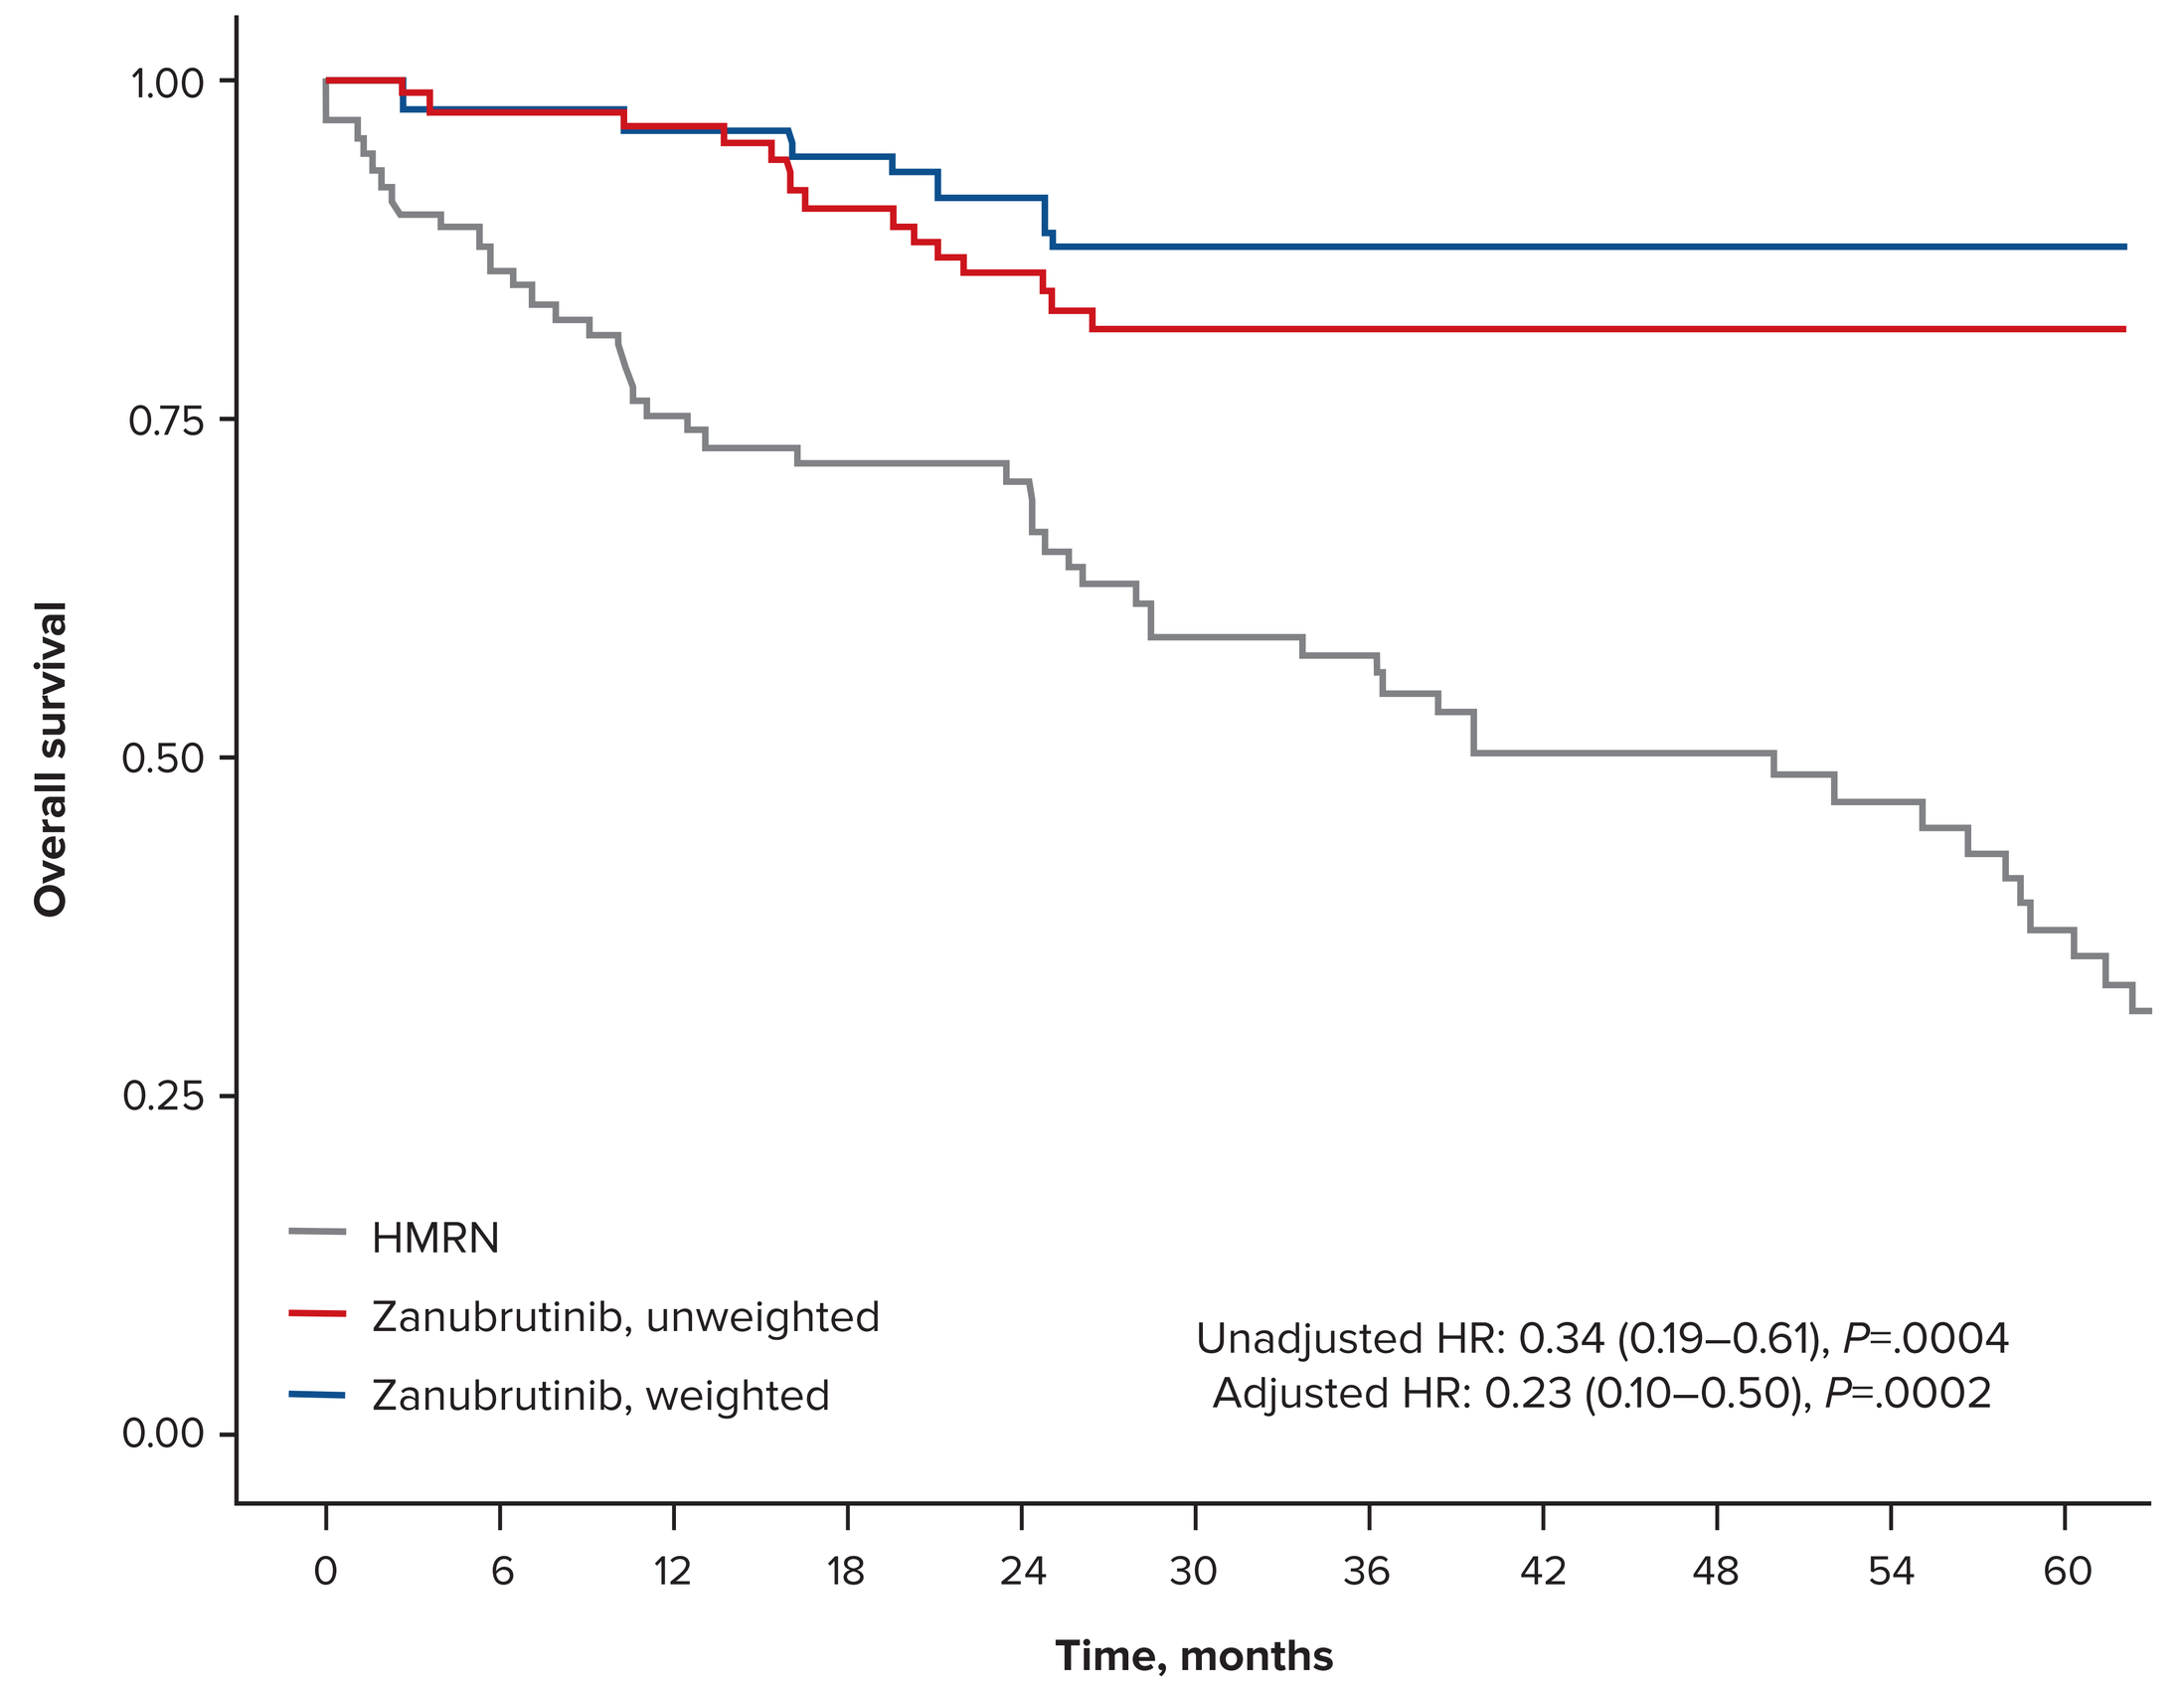
Overall survival
1.00
0.75
0.50
0.25
0.00
HMRN
Zanubrutinib, unweighted
Zanubrutinib, weighted
Unadjusted HR: 0.34 (0.19–0.61), P=.0004
Adjusted HR: 0.23 (0.10–0.50), P=.0002
0 6 12 18 24 30 36 42 48 54 60
Time, months

Number at risk

90	77	64	59	56	46	42	34	30	25	19
86	83	80	74	69	36	14	8	4	4	2
38	37	36	33	30	16	5	3	3	3	1

Values in parentheses are 95% CIs.

CONCLUSION

- MAIC results suggest that zanubrutinib can be considered an effective alternative to CIT, immunotherapy, or chemotherapy, adding to a body of evidence informing the relative efficacy of treatment options in patients with R/R MZL

DISCUSSION

- Given a limited number of large clinical trials investigating repeat CIT, immunotherapy, or chemo in R/R MZL settings, an MAIC utilizing real-world evidence can be considered as appropriate and informative
- To increase comparability of the populations, this study identified a subset of patients from the HMRN cohort most aligned with the inclusion and exclusion criteria of the zanubrutinib trials
- While patient factors considered to have the largest impact on prognosis were adjusted for in the analyses, residual bias from other unbalanced confounding variables remains a possibility
- The precision of MAIC estimates is contingent on the degree of patient overlap across the included studies; this can be observed in the adjusted models, whereby a large reduction from the original sample size was observed
- No comparison of safety outcomes was performed as these were not available from the HMRN registry
 - In general, rituximab is known to be a well-tolerated treatment in lymphoma and bendamustine plus rituximab is considered the most toxic of the treatment basket by clinicians
 - The adverse events from zanubrutinib were predominantly mild in nature with only temporary interruptions in treatment and with no treatment-related discontinuation, dose reduction, or fatality, with the exception of one patient from BGB-3111-AU-003

REFERENCES

1. Thieblemont C, et al. *J Clin Oncol.* 2023;14(16_suppl):e19527.
2. Thieblemont C, et al. *Hemasphere*. 2023;7(suppl):e10013c4.
3. Opat S, et al. *Clin Cancer Res*. 2021;27(23):6323-6332.
4. Phillips T, et al. *Blood Adv*. 2022;6(11):3472-3479.
5. HMRN. The Clinical Management and Outcome of Marginal Zone Lymphoma (MZL) with a Focus on Relapsed/Refractory Disease: A Report from the Haematological Malignancy Research Network. Updated February 24, 2024. Accessed March 20, 2024. https://hmrn.org/publications/reports

DISCLOSURES

RW: Honoraria: Janssen, AstraZeneca, AbbVie, BeiGene; Advisory Board: Janssen, AstraZeneca, SecuraBio, AbbVie, BeiGene. **KW, LM, and SM**: Employment: BeiGene. **CS, IZ, and SK**: Employment: PRECISIONheor; Research funding: BeiGene.

ACKNOWLEDGEMENTS

The authors thank the patients who participated in the MAGNOLIA and BGB-3111-AU-003 studies.

The authors also thank HMRN for granting access to valuable registry data, which contributed to the findings presented in this abstract.

This study was sponsored by BeiGene. Editorial assistance was provided by PRECISIONheor, and supported by BeiGene.

Presented at EHA 2024; June 13-16, 2024; Madrid, Spain/Virtual Conference