Efficacy and Safety of Zanubrutinib Versus Venetoclax+Ibrutinib in Treatment-Naïve Chronic Lymphocytic Leukemia: A Matching-Adjusted Indirect Comparison

P702

Talha Munir,[1] Leyla Mohseninejad,[2] Pal Rakonczai,[3] Sheng Xu,[4] Keri Yang,[5] Aileen Cohen,[5] Remus Vezan,[5] Nicolas Martinez-Calle[6]

[1]Leeds Teaching Hospitals NHS Trust, Leeds, UK; [2]BeiGene Netherlands B.V., Schiphol, The Netherlands; [3]Evidera, Budapest, Hungary; [4]BeiGene (Shanghai) Co, Ltd, Shanghai, China; [5]BeiGene USA, Inc, San Mateo, CA, USA; [6]Nottingham University Hospital, Nottingham, UK

INTRODUCTION

- Zanubrutinib is a highly effective next-generation Bruton tyrosine kinase inhibitor (BTKi) approved for the treatment of chronic lymphocytic leukemia (CLL). The phase 3 SEQUOIA study[1,2] evaluated the efficacy and safety of zanubrutinib in treatment-naïve (TN) patients with CLL with no del(17p) mutations (Arm A, zanubrutinib; Arm B, bendamustine+rituximab), and with del(17p) mutations (Arm C, zanubrutinib)
- The combination of fixed-duration B-cell lymphoma-2 inhibitor (BCL2i) venetoclax plus the BTKi ibrutinib (V+I) was approved by the European Medicine Agency for the treatment of TN CLL. V+I has been evaluated in the phase 3 GLOW study[3] including older patients and/or those with comorbidities and excluding the del(17p)/*TP53* population, and in the phase 2 CAPTIVATE study including younger/fitter patients and no restrictions on del(17p) and *TP53*[4]
- As no head-to-head clinical studies comparing zanubrutinib versus V+I exist, an indirect comparison was conducted to evaluate the relative efficacy and safety of the 2 treatments

OBJECTIVE

- The objective of this study was to conduct a matching-adjusted indirect comparison (MAIC) between zanubrutinib and V+I, and reduce potential bias due to differences in study populations

METHODS

- Individual patient-level data (IPD) from SEQUOIA was matched against the aggregate data from GLOW and CAPTIVATE
- Due to the lack of common control arms linking SEQUOIA with GLOW or CAPTIVATE, 2 unanchored MAICs were conducted
 - In MAIC-1, the zanubrutinib population from Cohort 1 in SEQUOIA (Arm A) was reweighted to match the key characteristics of the V+I population in GLOW (**Figure 1**)

Figure 1. MAIC-1

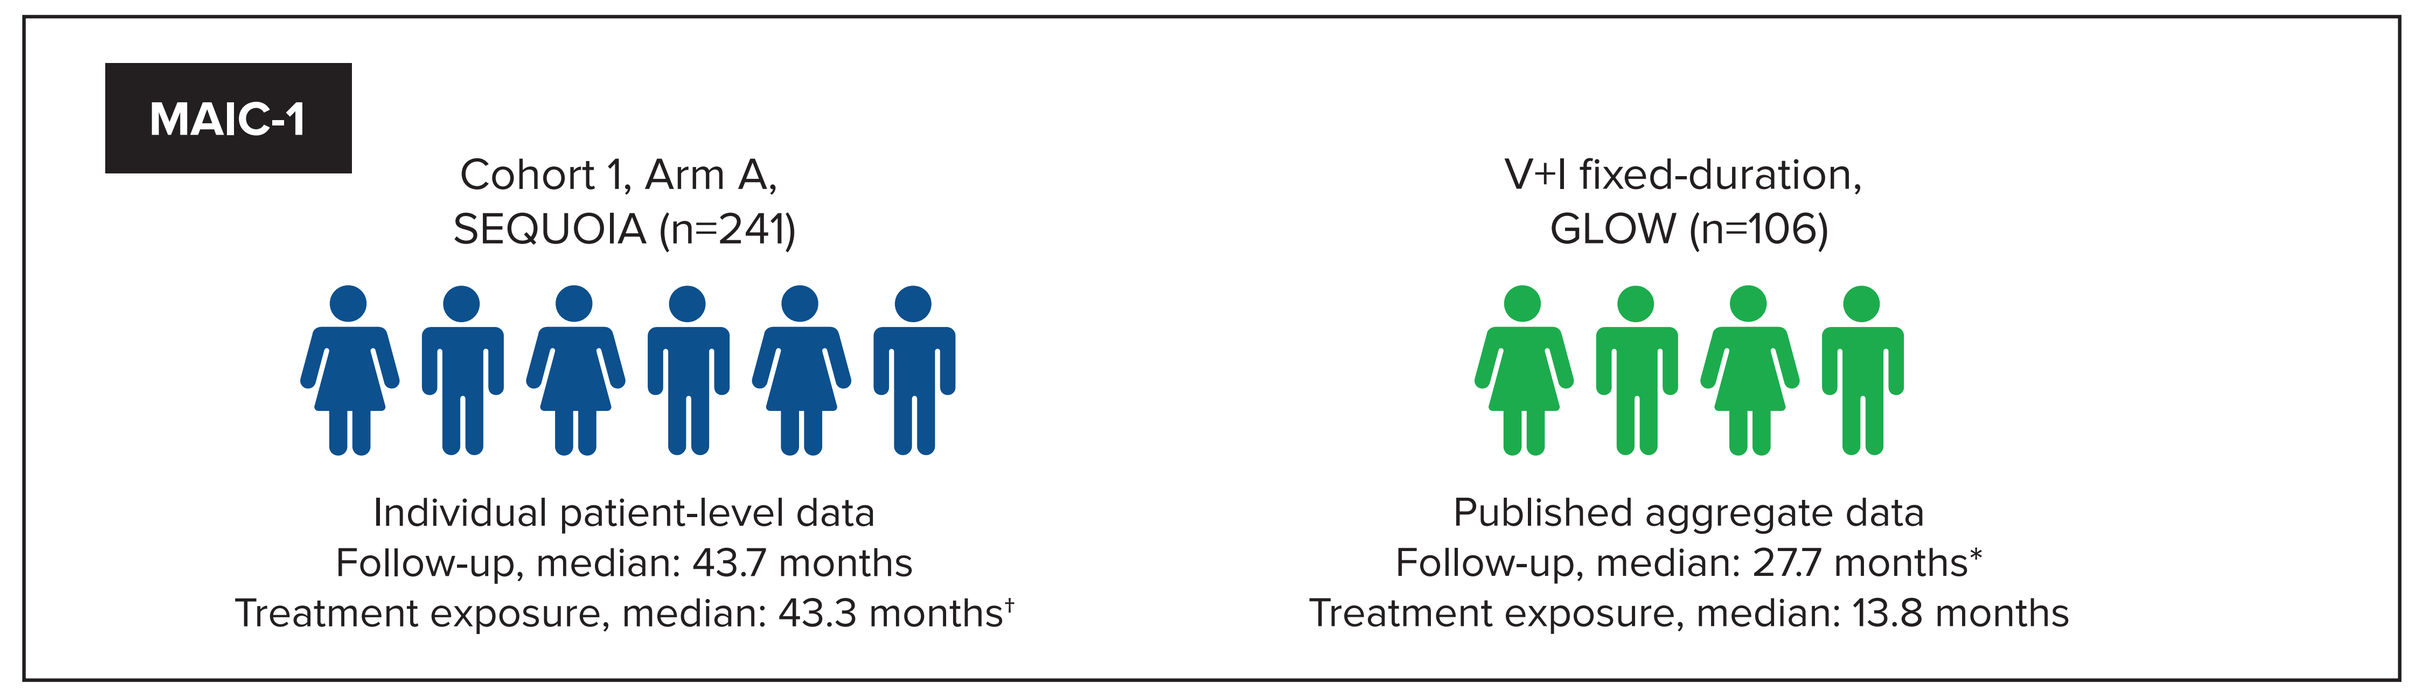
MAIC-1
Cohort 1, Arm A,
SEQUOIA (n=241)
Individual patient-level data
Follow-up, median: 43.7 months
Treatment exposure, median: 43.3 months†
V+I fixed-duration,
GLOW (n=106)
Published aggregate data
Follow-up, median: 27.7 months*
Treatment exposure, median: 13.8 months

† One patient who did not receive zanubrutinib treatment was excluded in the safety analysis.
* Median follow-up is not reported separately for V+I in GLOW, but only for pooled arms.

 - In MAIC-2, the pooled zanubrutinib population from Cohort 1 and 2 in SEQUOIA (Arm A+C) was reweighted to match the key characteristics of the V+I population in CAPTIVATE (**Figure 2**)

Figure 2. MAIC-2

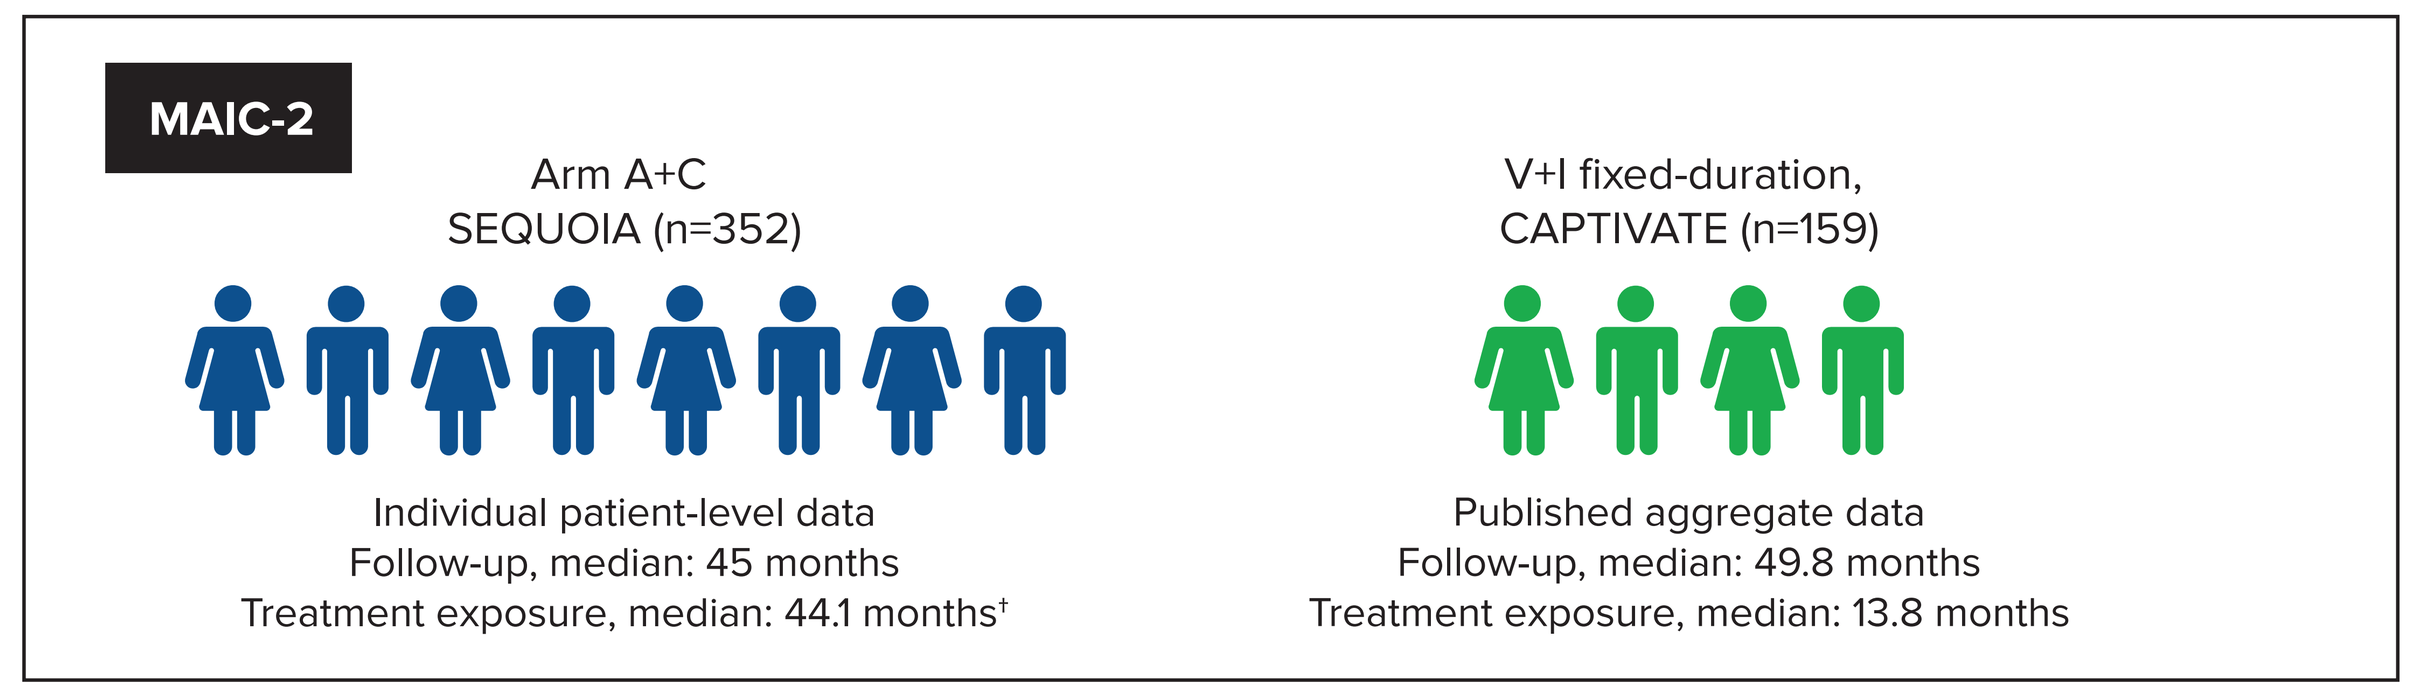
MAIC-2
Arm A+C
SEQUOIA (n=352)
Individual patient-level data
Follow-up, median: 45 months
Treatment exposure, median: 44.1 months†
V+I fixed-duration,
CAPTIVATE (n=159)
Published aggregate data
Follow-up, median: 49.8 months
Treatment exposure, median: 13.8 months

† One patient who did not receive zanubrutinib treatment was excluded in the safety analysis.

- Adjustments for all key population characteristics identified as potential treatment effect modifiers or prognostic factors including age, sex, geographic region, Eastern Cooperative Oncology Group Performance Status (ECOG PS), cancer type, CLL stage, genomic mutation status, bulky disease, beta$_2$-microglobulin, complex karyotype, time from diagnosis, Cumulative Illness Rating Score (CIRS), and creatinine clearance were considered based on availability across the studies, clinical relevance, and magnitude of imbalance across study populations
- Adjusting for all commonly available characteristics resulted in insufficiently low effective sample size (ESS); several simpler matching models were explored to find the most optimal matches. After the matching model was identified, balancing weights were derived for zanubrutinib patients in the SEQUOIA study
- Pseudo IPD for investigator-assessed progression-free survival (PFS-INV) in the V+I arms were reconstructed from the digitized Kaplan-Meier (KM) curves reported in the GLOW and CAPTIVATE publications using the algorithm by Guyot et al[5]
- Weighted Cox proportional hazard regression was used to derive relative treatment effect estimates for PFS-INV
- Given that matching variables mainly impact efficacy and not safety, the primary safety comparison did not apply weights used in the efficacy comparison; naïve comparison using logistic regression was carried out as the primary approach. Weighted logistic regression using weights from the efficacy comparison was carried out as part of the sensitivity analysis

RESULTS

Population Adjustment

- A matching model adjusting for all commonly available characteristics, except CIRS, including age, sex, geographic region, ECOG PS, cancer type, CLL stage, various genomic mutation status, bulky disease, beta2-microglobulin, time from diagnosis, and creatinine clearance level in the GLOW population was successfully fitted with ESS=152 (63% of Arm A in SEQUOIA) (**Table 1**). CIRS was excluded from the matching due to the large imbalance between SEQUOIA (27%) and GLOW (70%) populations, which would lead to a drastic drop in ESS. Adding CIRS was explored as part of the sensitivity analysis and results are presented in the sensitivity analysis section

Table 1. Baseline Characteristics of Zanubrutinib Arm in SEQUOIA and V+I Arm in GLOW

Population Characteristics	Included in Matching	V+I in GLOW n=106	Zanubrutinib in SEQUOIA Cohort 1 Prematching, n=241	Zanubrutinib in SEQUOIA Cohort 1 Postmatching, ESS=152
Median age, years	✓	71	70	71
Male, %	✓	55.7%	63.9%	55.7%
Geographic region, North America, or Europe, %	✓	86.8%	86.3%	86.8%
ECOG PS = 0, %	✓	33.0%	45.6%	33.0%
SLL (vs CLL), %	✓	9.4%	8.3%	9.4%
Binet stage C, %	✓	40.6%	29.0%	40.6%
Mutated IGHV, %	✓	32.9%	47.2%	32.9%
Del(17p), %	✓	0.0%	0.8%	0.0%
Del(11q), %	✓	18.9%	17.8%	18.9%
TP53 mutation, %	✓	6.6%	6.5%	6.6%
Bulky disease, LD in cm, ≥5, %	✓	39.0%	28.6%	39.0%
Beta$_2$-microglobulin >3.5 mg/L, %	✓	69.8%	57.3%	69.8%
Median time from diagnosis, months	✓	35.8	31	36
Median creatinine clearance, mL/min	✓	66.5	71	66
CIRS >6, %	x	69.8%	27.1%	30.9%

CIRS, Cumulative Illness Rating Score; IGHV, immunoglobulin heavy-chain variable; LD, longest diameter; SLL, small lymphocytic leukemia.

- Compared to SEQUOIA, the CAPTIVATE population was significantly younger (median age, SEQUOIA: 70 years; CAPTIVATE: 60 years) and had significantly better heath state (proportion of patients with lowest ECOG PS was 69.2% vs 43.8%) (**Table 2**). Therefore, a matching model adjusting for age, sex, ECOG PS, cancer type, CLL stage, and various genomic mutation status was fitted with rather low ESS=51, only 14.5% of the sample size of Arm A+C population in SEQUOIA
- Further adjustment for bulky disease and complex karyotype was not feasible due to their impact on the ESS
- Baseline characteristics related to beta$_2$-microglobulin and geographic region were not available in CAPTIVATE for matching

Table 2. Matching and Baseline Characteristics Between CAPTIVATE and SEQUOIA Cohort 1 and 2

Population Characteristics	Included in Matching	V+I in CAPTIVATE n=159	Zanubrutinib in SEQUOIA Cohort 1 and 2 Prematching n=352	Zanubrutinib in SEQUOIA Cohort 1 and 2 Postmatching ESS=51
Median age, years	✓	60	70	60
Maximum age, years	✓	71	87	71
Male, %	✓	67.0%	66.2%	67.0%
ECOG PS = 0, %	✓	69.2%	43.8%	69.2%
SLL (vs CLL), %	✓	8.0%	8.8%	8.0%
Rai stage III-IV,† %	✓	28.0%	31.0%	28.0%
Mutated IGHV, %	✓	42.6%	43.8%	42.6%
Del(17p), %	✓	12.7%	31.8%	12.7%
Del(11q)*, %	✓	17.7%	11.6%	17.7%
Trisomy 12*, %	✓	14.6%	12.5%	14.6%
Normal*, %	✓	20.9%	15.9%	20.9%
Del(13q)*, %	✓	34.1%	28.2%	34.1%
TP53 mutation, %	✓	10.3%	18.2%	10.3%
Bulky disease, LD in cm, ≥5, %	✓	30.2%	32.1%	25.6%
Complex karyotype ≥3 abnormalities, %	x	23.3%	22.3%	16.0%

† Since Rai stage was not reported in SEQUOIA, Binet C was used as a proxy for Rai III-IV. * As per hierarchical classification.
LD, longest diameter; V+I, venetoclax + ibrutinib.

PFS-INV

- Both naïve and population-adjusted comparison of PFS-INV for zanubrutinib in SEQUOIA (Arm A) vs V+I in GLOW indicated a trend for potential treatment benefit in favor of zanubrutinib with HR=0.71 (P=.2578) and HR=0.84 (P=.5977), respectively (**Figure 3**), however, no statistical significance was observed
- While in the naïve comparison of PFS-INV for zanubrutinib in SEQUOIA (Arm A+C) vs V+I in CAPTIVATE with HR=1.00 (P=.9874) indicated that treatment effects were similar, the population-adjusted estimate HR=0.78 (P=.5099) indicated a trend for potential treatment benefit in favor of zanubrutinib vs V+I (**Figure 4**). No statistical significance was observed
- Due to the relatively low sample sizes in V+I arms in GLOW and CAPTIVATE and the relatively low overlap in population characteristics across SEQUOIA and the V+I studies, both naïve and population-adjusted relative treatment effect estimates had large uncertainty. Therefore, no statistically significant differences in PFS could be demonstrated for zanubrutinib vs V+I
- As shown in the comparison of postmatching zanubrutinib vs V+I (CAPTIVATE) (**Figure 4**), the PFS-INV KM curves overlap over the first 9 months, then the V+I curve is above the zanubrutinib curve up to 28 months when the curves start overlapping again. In addition, there is notable declination in the V+I curve at around 40 months that could be partly explained by the fact V+I is fixed-duration, while zanubrutinib is a continuous treatment with no declination related to any specific timepoint during the observed period. The difference observed at the tail of the 2 curves leads to a HR estimate indicating a numerical benefit in favor of zanubrutinib
 - Longer follow-up would be required to make a more informed conclusion, as well as adjustment for the misalignment in the assessment schedules between the 2 studies

Figure 3. MAIC-1 PFS-INV

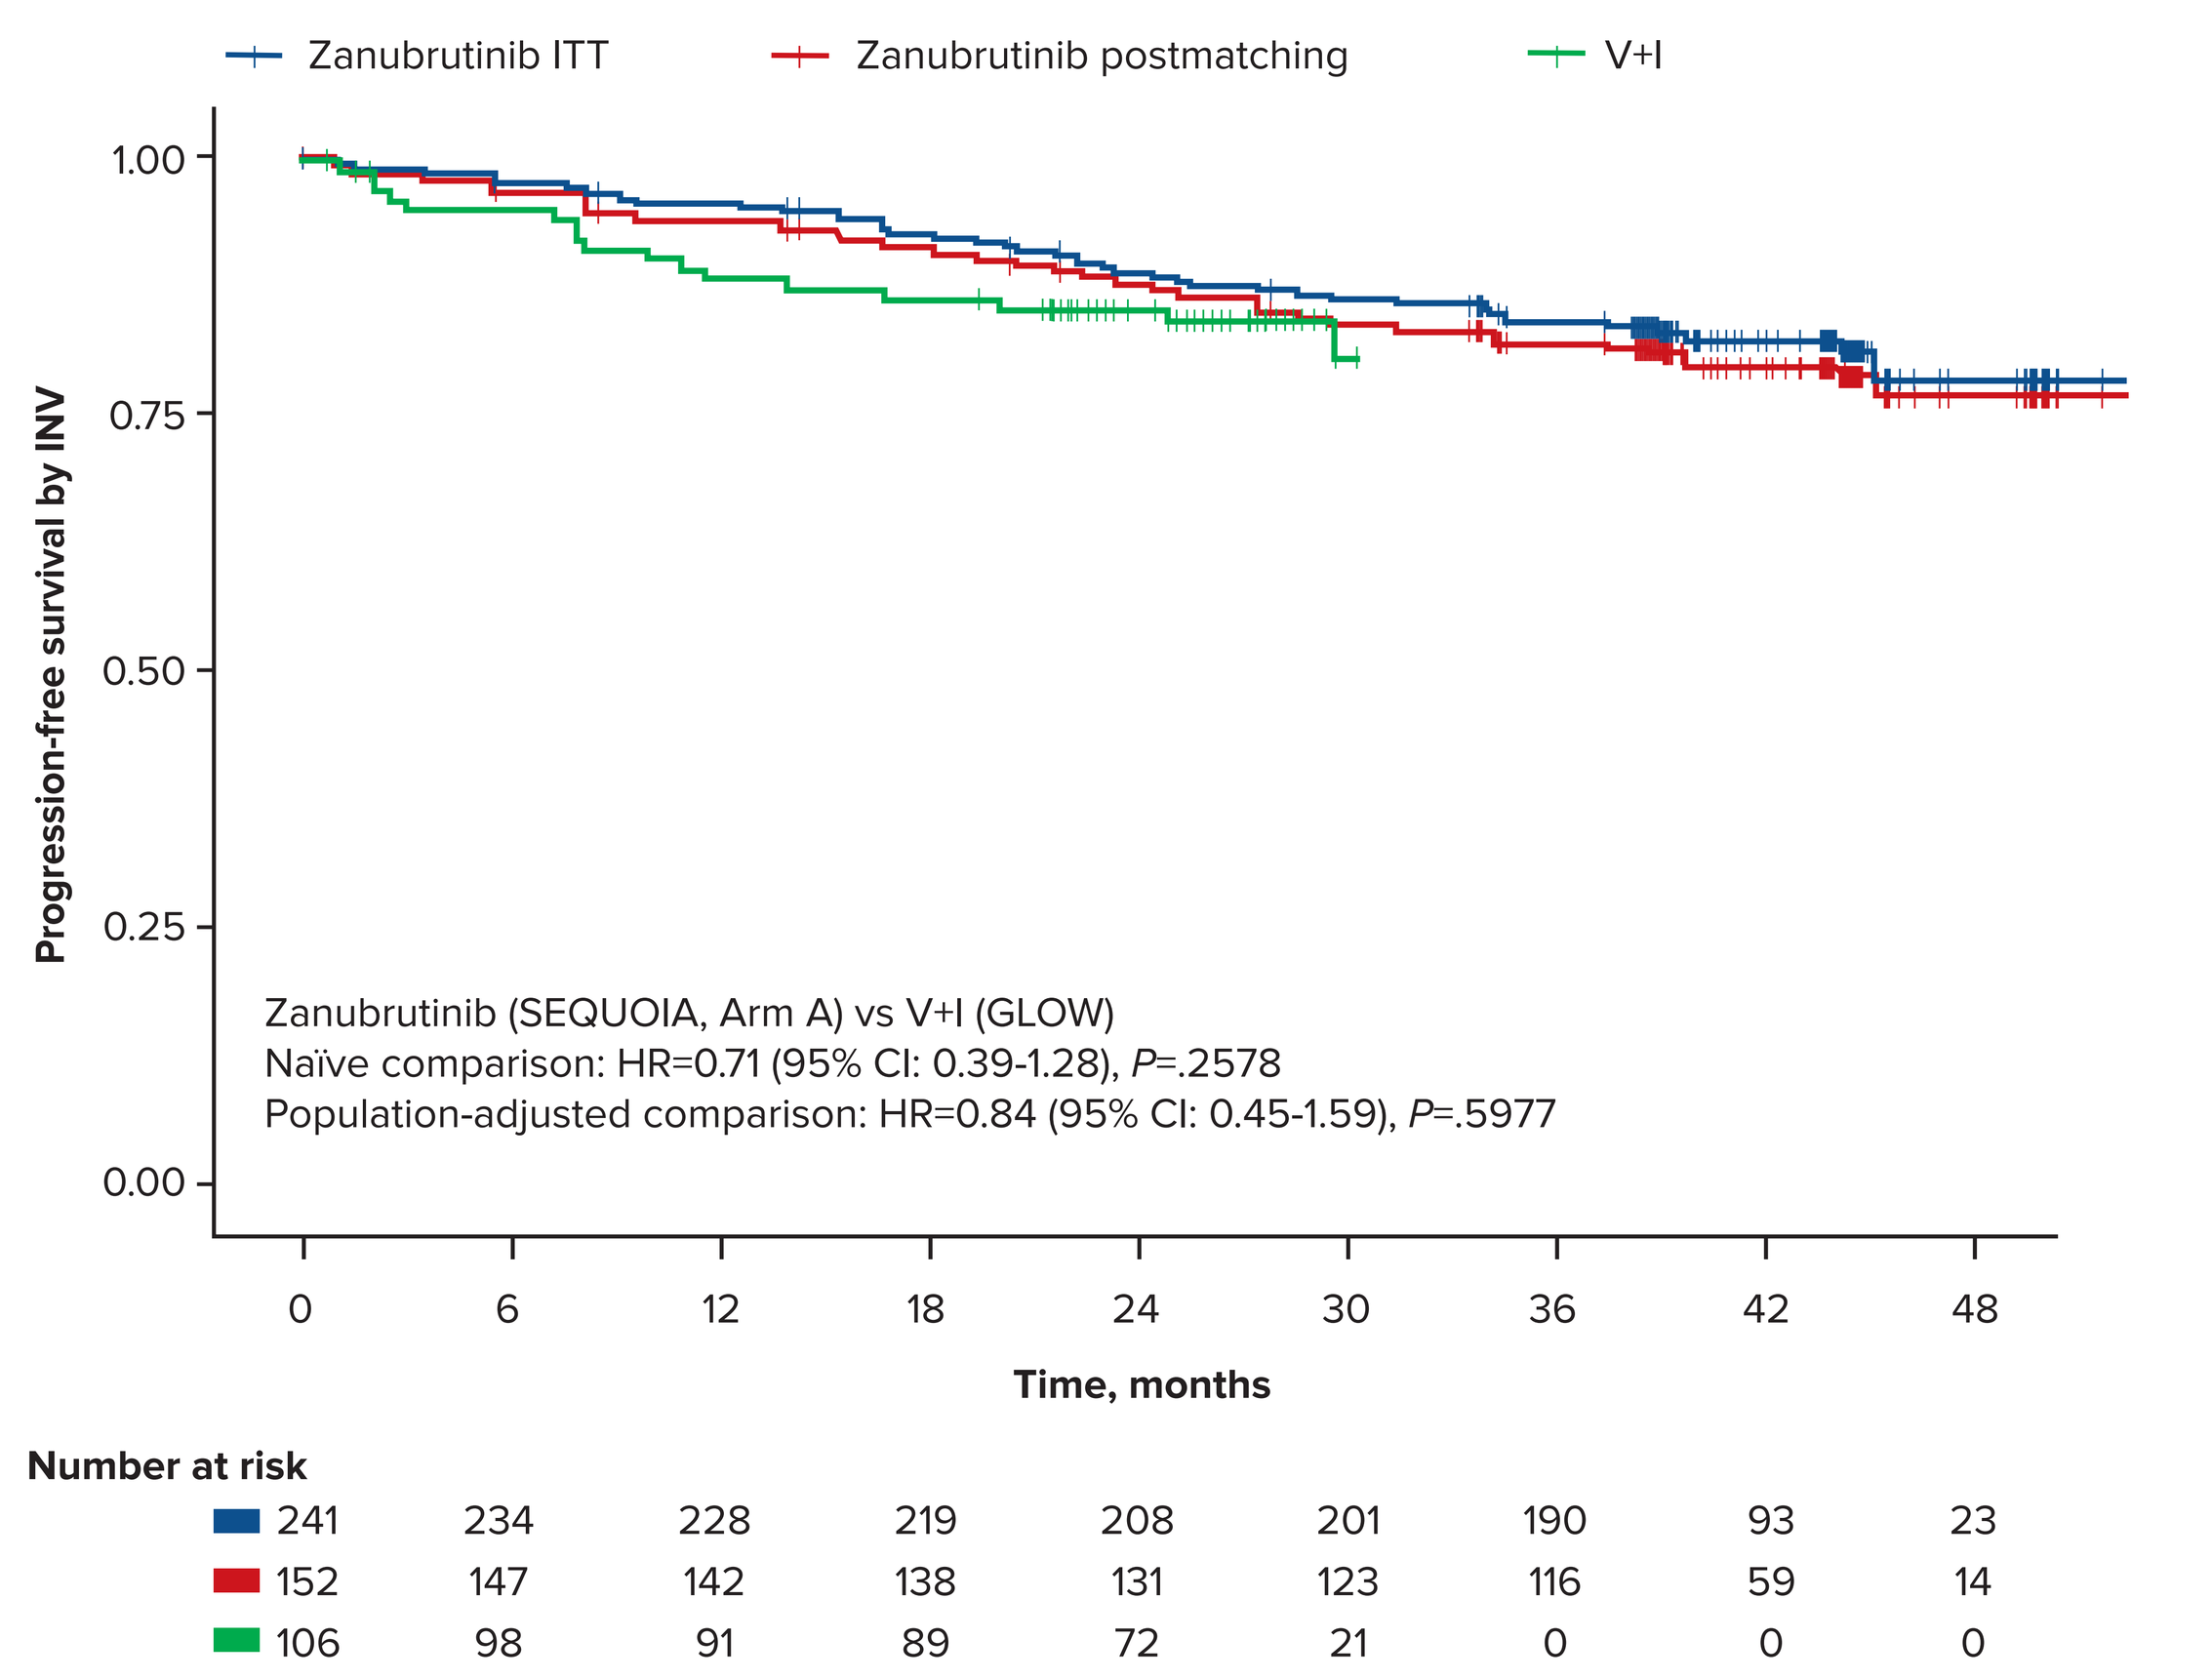
Zanubrutinib ITT
Zanubrutinib postmatching
V+I
Progression-free survival by INV
1.00
0.75
0.50
0.25
0.00
Zanubrutinib (SEQUOIA, Arm A) vs V+I (GLOW)
Naïve comparison: HR=0.71 (95% CI: 0.39-1.28), P=.2578
Population-adjusted comparison: HR=0.84 (95% CI: 0.45-1.59), P=.5977
0 6 12 18 24 30 36 42 48
Time, months
Number at risk
241 234 228 219 208 201 190 93 23
152 147 142 138 131 123 116 59 14
106 98 91 89 72 21 0 0 0

Figure 4. MAIC-2 PFS-INV

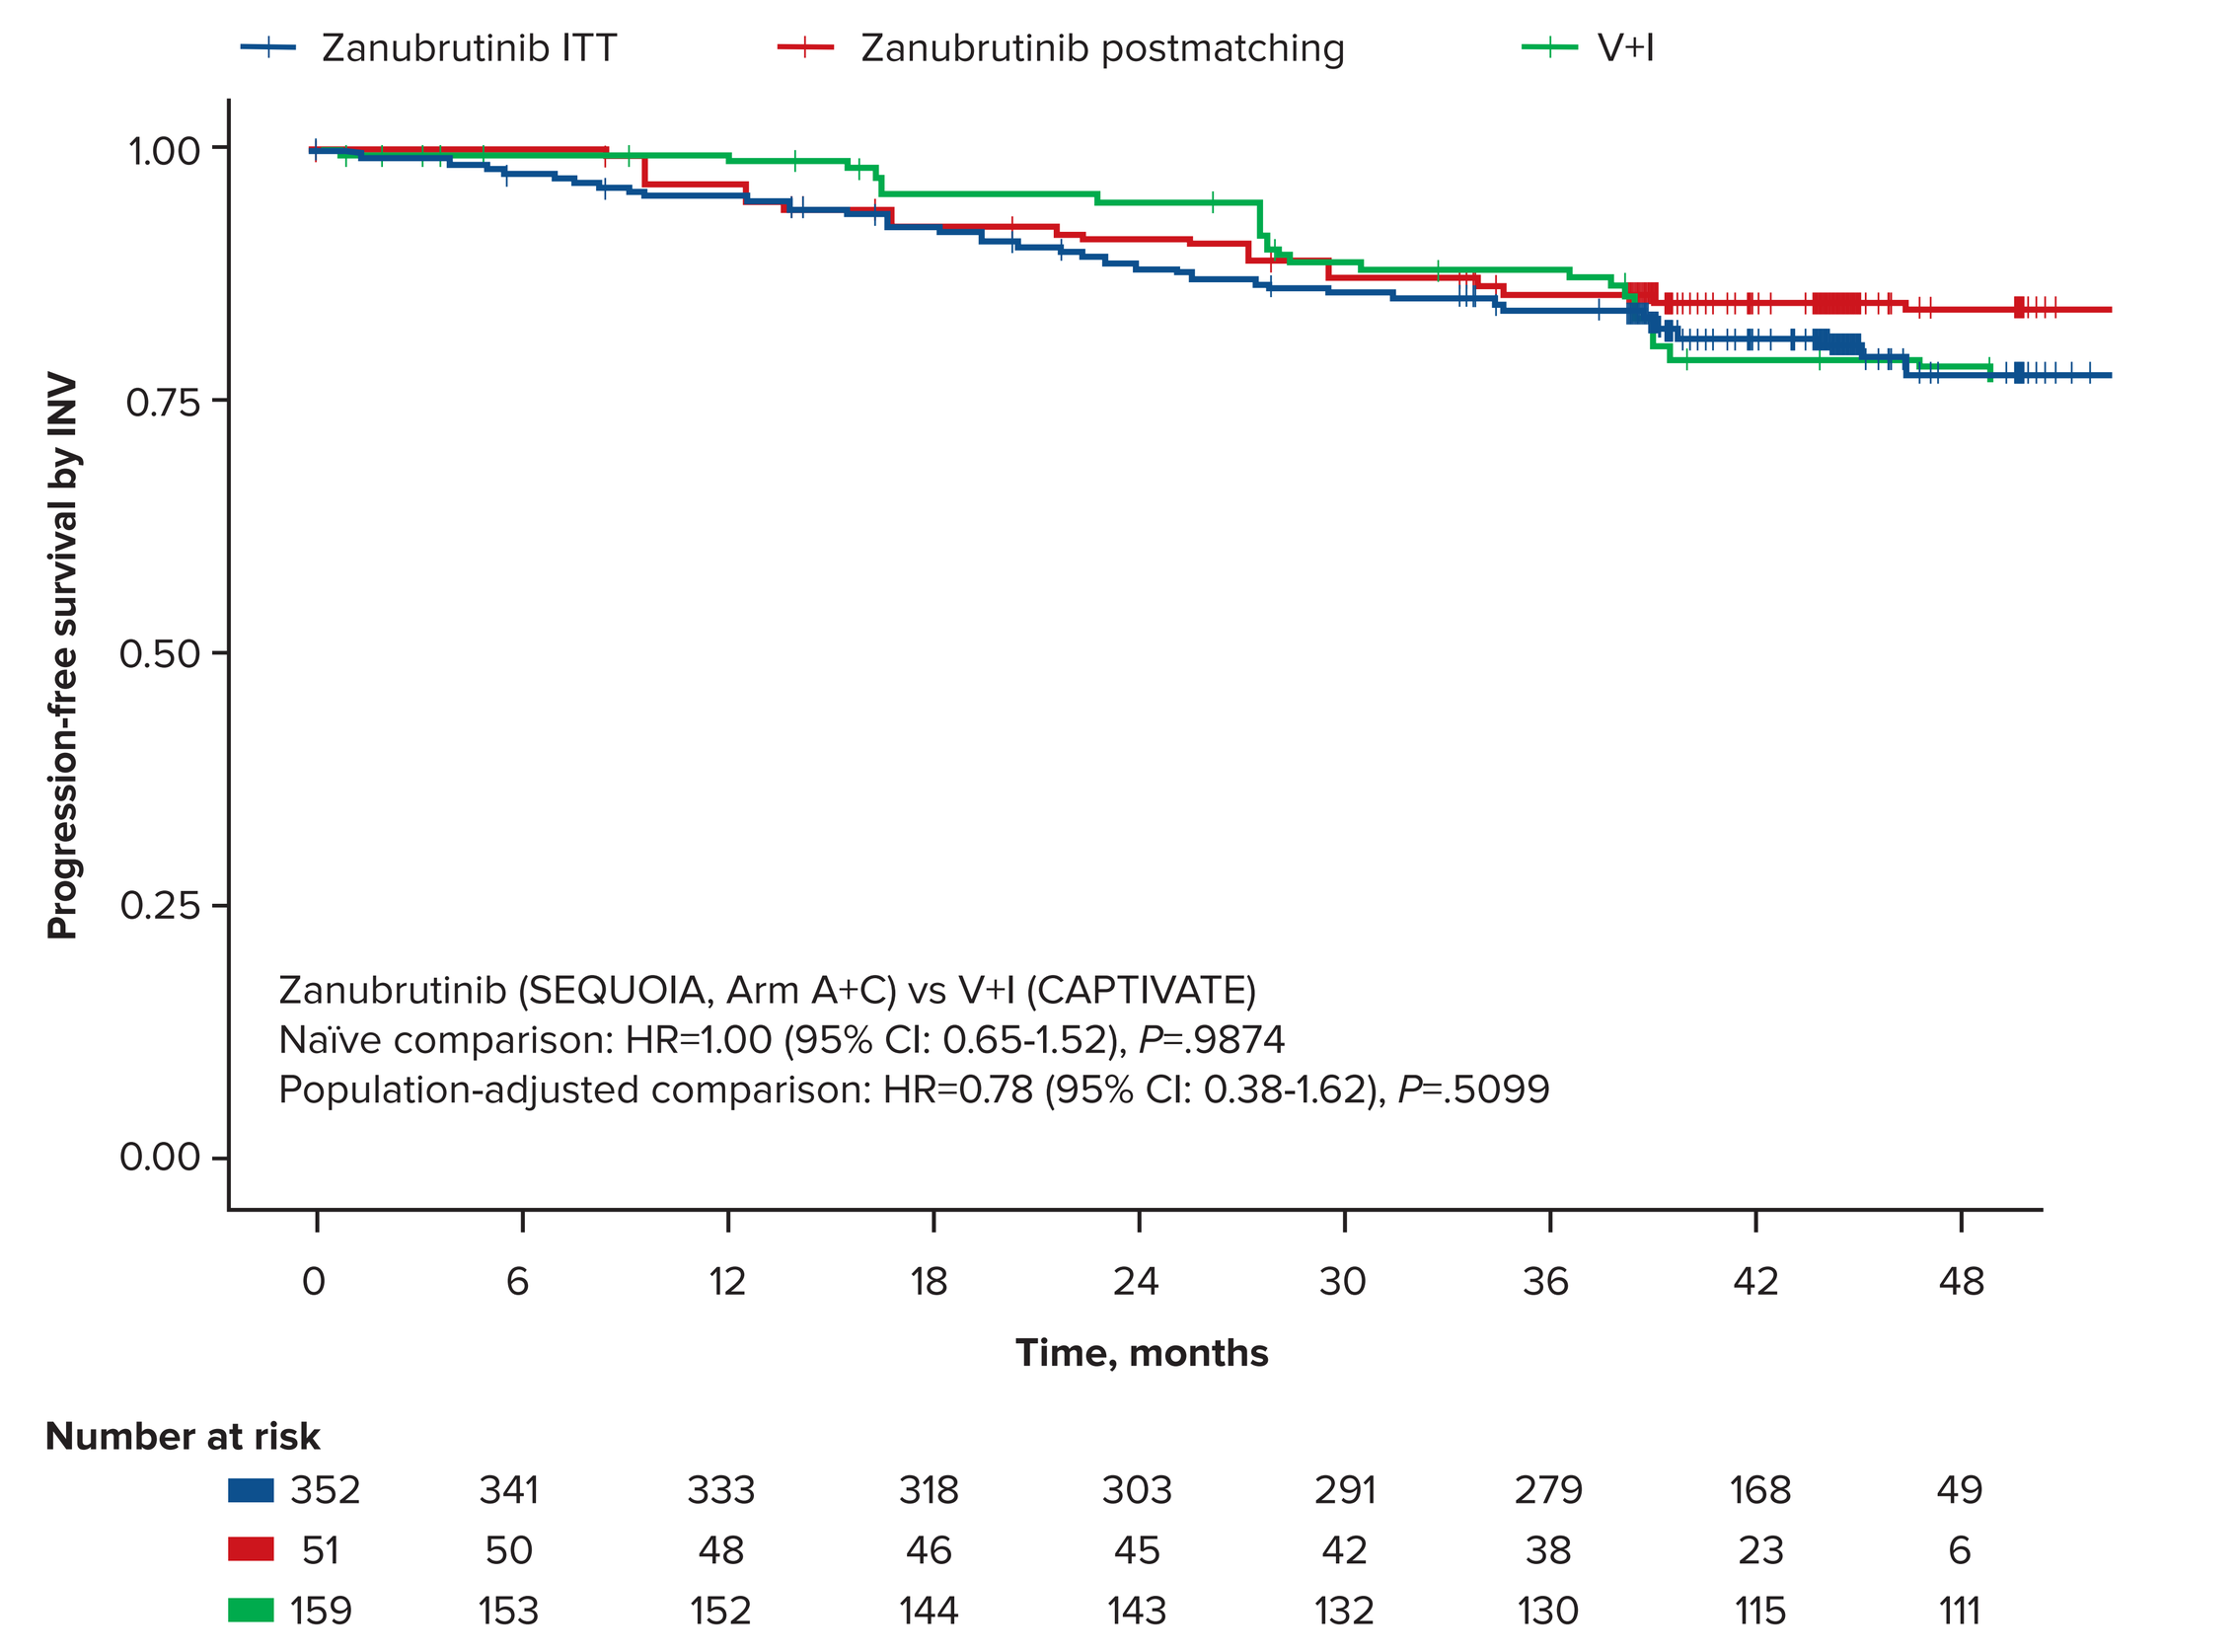
Zanubrutinib ITT
Zanubrutinib postmatching
V+I
Progression-free survival by INV
1.00
0.75
0.50
0.25
0.00
Zanubrutinib (SEQUOIA, Arm A+C) vs V+I (CAPTIVATE)
Naïve comparison: HR=1.00 (95% CI: 0.65-1.52), P=.9874
Population-adjusted comparison: HR=0.78 (95% CI: 0.38-1.62), P=.5099
0 6 12 18 24 30 36 42 48
Time, months
Number at risk
352 341 333 318 303 291 279 168 49
51 50 48 46 45 42 38 23 6
159 153 152 144 143 132 130 115 111

CONCLUSIONS

- No statistically significant differences in PFS could be demonstrated for zanubrutinib vs V+I. However, the adjusted estimates showed a potential trend for treatment benefit in favor of zanubrutinib vs V+I
- Zanubrutinib showed a significantly better safety profile vs V+I, despite longer treatment exposure (median 43-44 months vs 13.8 months in both V+I studies)
- The observed safety differences between zanubrutinib and V+I suggest considerable impact on patients' quality of life, which should be considered at the time of treatment decision-making in TN CLL

- Comparing against CAPTIVATE, including adjustment for bulky disease instead of maximum age in the matching model (**Table 2**), resulted in ESS=55 and HR=0.83 (95% CI: 0.43-1.61, P=.5869). Similarly, including adjustment for complex karyotype resulted in ESS=32 and HR=0.78 (95% CI: 0.33-1.87, P=.5834)
 - To increase ESS, a simplified model adjusting for limited set of population characteristics was further explored. This exploratory model included adjustments only for IGHV, del(17p) and del(11q) mutation status, CLL stage, and age, and resulted in a considerably higher ESS of 91 and HR=0.66 (95% CI: 0.34-1.26, P=.2047)

Safety Outcomes

- Significantly lower incidence was indicated for zanubrutinib vs V+I in both GLOW and CAPTIVATE studies with regards to multiple AEs (**Figure 5**)
- Notably, zanubrutinib usage led to significantly lower proportions of diarrhea, neutropenia, nausea, anemia, atrial fibrillation, decreased appetite, and arthralgia

Sensitivity Analysis (safety)

- Sensitivity analyses exploring the impact of using the same matching factors from the efficacy comparison for the safety comparison demonstrated results that were consistent with the unadjusted safety comparison (see appendix in QR code)

Figure 5. Adverse Events of any Grade or Grade 3+ by Preferred Term with a *P* value <.2 in the Naïve Safety Comparison

Adverse Events	Comparator Study	V+I (Events/N)	Zanubrutinib (Events/N)	Naïve OR (95% CI)	*P* Value
Diarrhea, any grade	GLOW	54/106	41/240	0.20 (0.12–0.33)	<.001
	CAPTIVATE	99/159	63/351	0.13 (0.09–0.20)	<.001
Diarrhea, grade 3+	GLOW	11/106	4/240	0.15 (0.05–0.47)	.001
Neutropenia*, any grade	GLOW	44/106	32/240	0.22 (0.13–0.37)	<.001
	CAPTIVATE	66/159	45/351	0.21 (0.13–0.32)	<.001
Neutropenia*, grade 3+	GLOW	37/106	24/240	0.21 (0.12–0.37)	<.001
	CAPTIVATE	52/159	36/351	0.24 (0.15–0.38)	<.001
Nausea, any grade	GLOW	28/106	31/240	0.41 (0.23–0.73)	.003
	CAPTIVATE	68/159	51/351	0.23 (0.15–0.35)	<.001
Anemia, any grade	GLOW	19/106	16/240	0.33 (0.16–0.67)	.002
UTI, any grade	GLOW	17/106	24/240	0.58 (0.30–1.14)	.112
Edema peripheral, any grade	GLOW	16/106	23/240	0.60 (0.30–1.18)	.138
Atrial fibrillation, any grade	GLOW	15/106	10/240	0.26 (0.11–0.61)	.002
Atrial fibrillation, grade 3+	GLOW	7/106	2/240	0.12 (0.02–0.58)	.009
Decreased appetite, any grade	GLOW	14/106	7/240	0.20 (0.08–0.50)	.001
Thrombocytopenia, any grade	GLOW	12/106	12/240	0.41 (0.18–0.95)	.038
Thrombocytopenia, grade 3+	GLOW	6/106	4/240	0.28 (0.08–1.02)	.054
Arthralgia, any grade	CAPTIVATE	53/159	63/351	0.44 (0.29–0.67)	<.001
Epistaxis, any grade	GLOW	12/106	13/240	0.45 (0.20–1.02)	.056
Cough, any grade	GLOW	9/106	35/240	1.84 (0.85–3.98)	.121
Hyponatremia, grade 3+	GLOW	6/106	5/240	0.35 (0.11–1.19)	.093

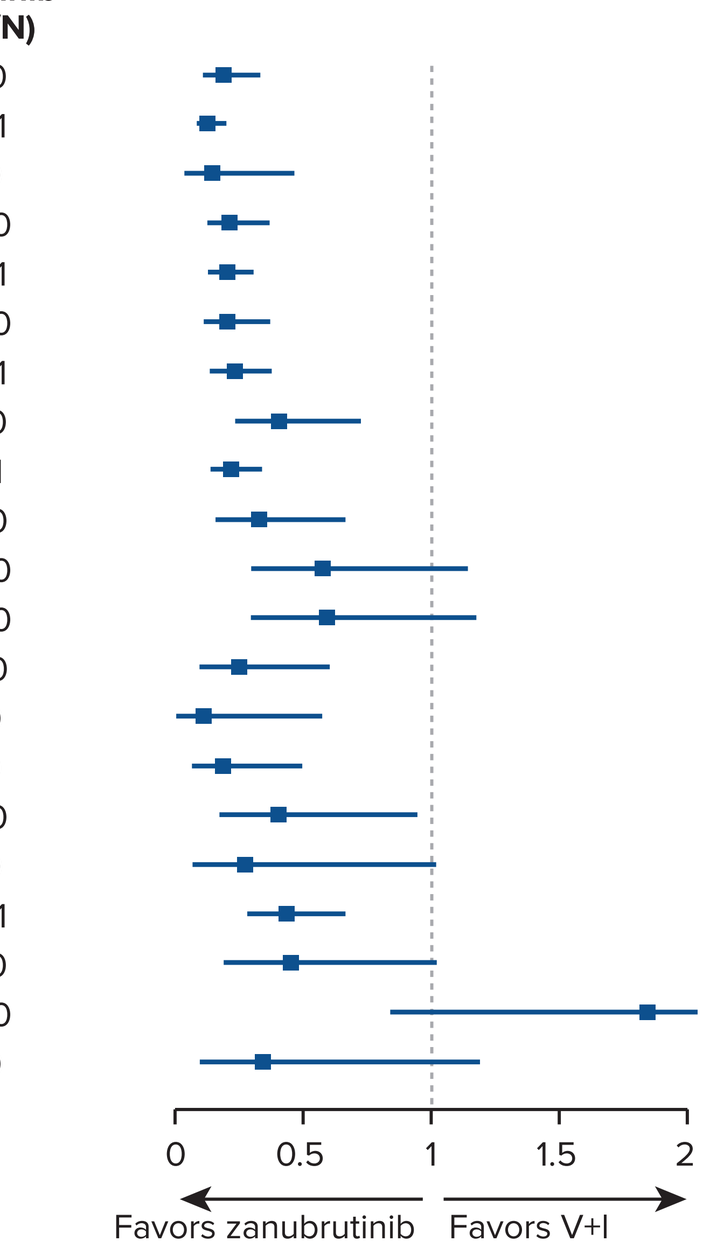
0
0.5
1
1.5
2
Favors zanubrutinib
Favors V+I

* In GLOW neutropenia indicates neutropenia and neutrophil count decreased.
UTI, urinary tract infection; V+I, venetoclax + ibrutinib.

LIMITATIONS

- In any MAIC, there is a potential for bias resulting from the strong assumption that cross-trial differences can be entirely explained by variables selected for matching
- Overall survival was not included in the MAIC analysis due to immaturity of data
- An important limitation of the safety comparison is that CAPTIVATE reported AEs only if they exceeded 30% in all grade or 5% in grade 3+. Therefore, the comparison of AEs with 10%-30% incidence in all grade was not possible unless their grade 3+ incidence exceeded 5%

REFERENCES

1. Tam CS, et al. *Lancet Oncol*. 2022;23:1031-1043.
2. Munir T, et al. Poster session presented at: EHA 2023; 2023 June 8-11; Frankfurt, Germany.
3. ClinicalTrials.gov identifier: NCT03462719. Updated 2023-03-28. https://clinicaltrials.gov/study/NCT03462719
4. ClinicalTrials.gov identifier: NCT02910583. Updated 2023-04-03. https://clinicaltrials.gov/study/NCT02910583
5. Guyot P, et al. *BMC Med Res Methodol*. 2012;12:9.

DISCLOSURES

TM: Research Funding: Janssen, AbbVie; Honoraria: AbbVie, Janssen, AstraZeneca, Eli Lilly, Novartis, Roche, Sobi, Alexion, BeiGene; Advisory Committees: AbbVie, Janssen, AstraZeneca, Eli Lilly, Novartis, Roche, Sobi, Alexion, BeiGene. **LM, SX, KY, AC, RV**: Employment and may hold stock options: Beigene. **PR**: Employment: Evidera. **NMC**: Honoraria: BeiGene, AbbVie, AstraZeneca; Consulting: BeiGene, AbbVie, Takeda; Travel support: AbbVie

ACKNOWLEDGEMENTS

This study was sponsored by BeiGene, Ltd. Editorial assistance was provided by Evidera, and supported by BeiGene.

Presented at EHA 2024; June 13-16, 2024; Madrid, Spain/Virtual Conference