Budget Impact Analysis of Zanubrutinib for Patients with Treatment-naïve Chronic Lymphocytic Leukemia or Small Lymphocytic Lymphoma in the United States

EE336

Sizhu Liu[1], Keri Yang[1], Ying Xiao[2], Ray Gani[2], Mei Xue[1], Boxiong Tang[1]

[1] BeiGene USA, Inc., San Mateo, CA, United States; [2] Evidera, London, United Kingdom

Background

- Chronic lymphocytic leukemia/small lymphocytic lymphoma (CLL/SLL) is an indolent lymphoproliferative disorder and is characterized by the progressive accumulation of monoclonal, small, mature-appearing CD5+ B-lymphocytes mainly within peripheral blood, bone marrow, lymph nodes, and secondary lymphoid organs.[1]
- Bruton's tyrosine kinase inhibitor (BTKi) plays a key role in B-cell receptor signaling. It introduced the paradigm change in the CLL/SLL treatment pathway and has challenged the role of chemo-immunotherapy (CIT).[2]
- A recent update to National Comprehensive Cancer Network (NCCN) clinical guidelines included zanubrutinib, a next-generation BTKi, as preferred treatment for treatment-naive (TN) CLL/SLL.[3] The efficacy and safety of zanubrutinib are examined in the randomized, phase 3 SEQUOIA trial (NCT03336333) comparing zanubrutinib to bendamustine plus rituximab (BR) in cohort 1 patients with previously untreated CLL/SLL and without del 17p mutation.[4]
- The objective of this study is to develop a budget impact model (BIM) and estimate the incremental costs associated with using zanubrutinib in the population from the US commercial and Medicare perspectives.

Methods

Model Design

- A Microsoft Excel-based BIM was developed to compare a "reference scenario" reflecting the current market scenario of treatment options (i.e., before zanubrutinib entry), with an "alternative scenario" which also included zanubrutinib for a proportion of patients based on its projected uptake in TN CLL/SLL patients (i.e., after zanubrutinib entry) over a three-year time horizon. (**Figure 1**)

Figure 1. Budget Impact Model Structure

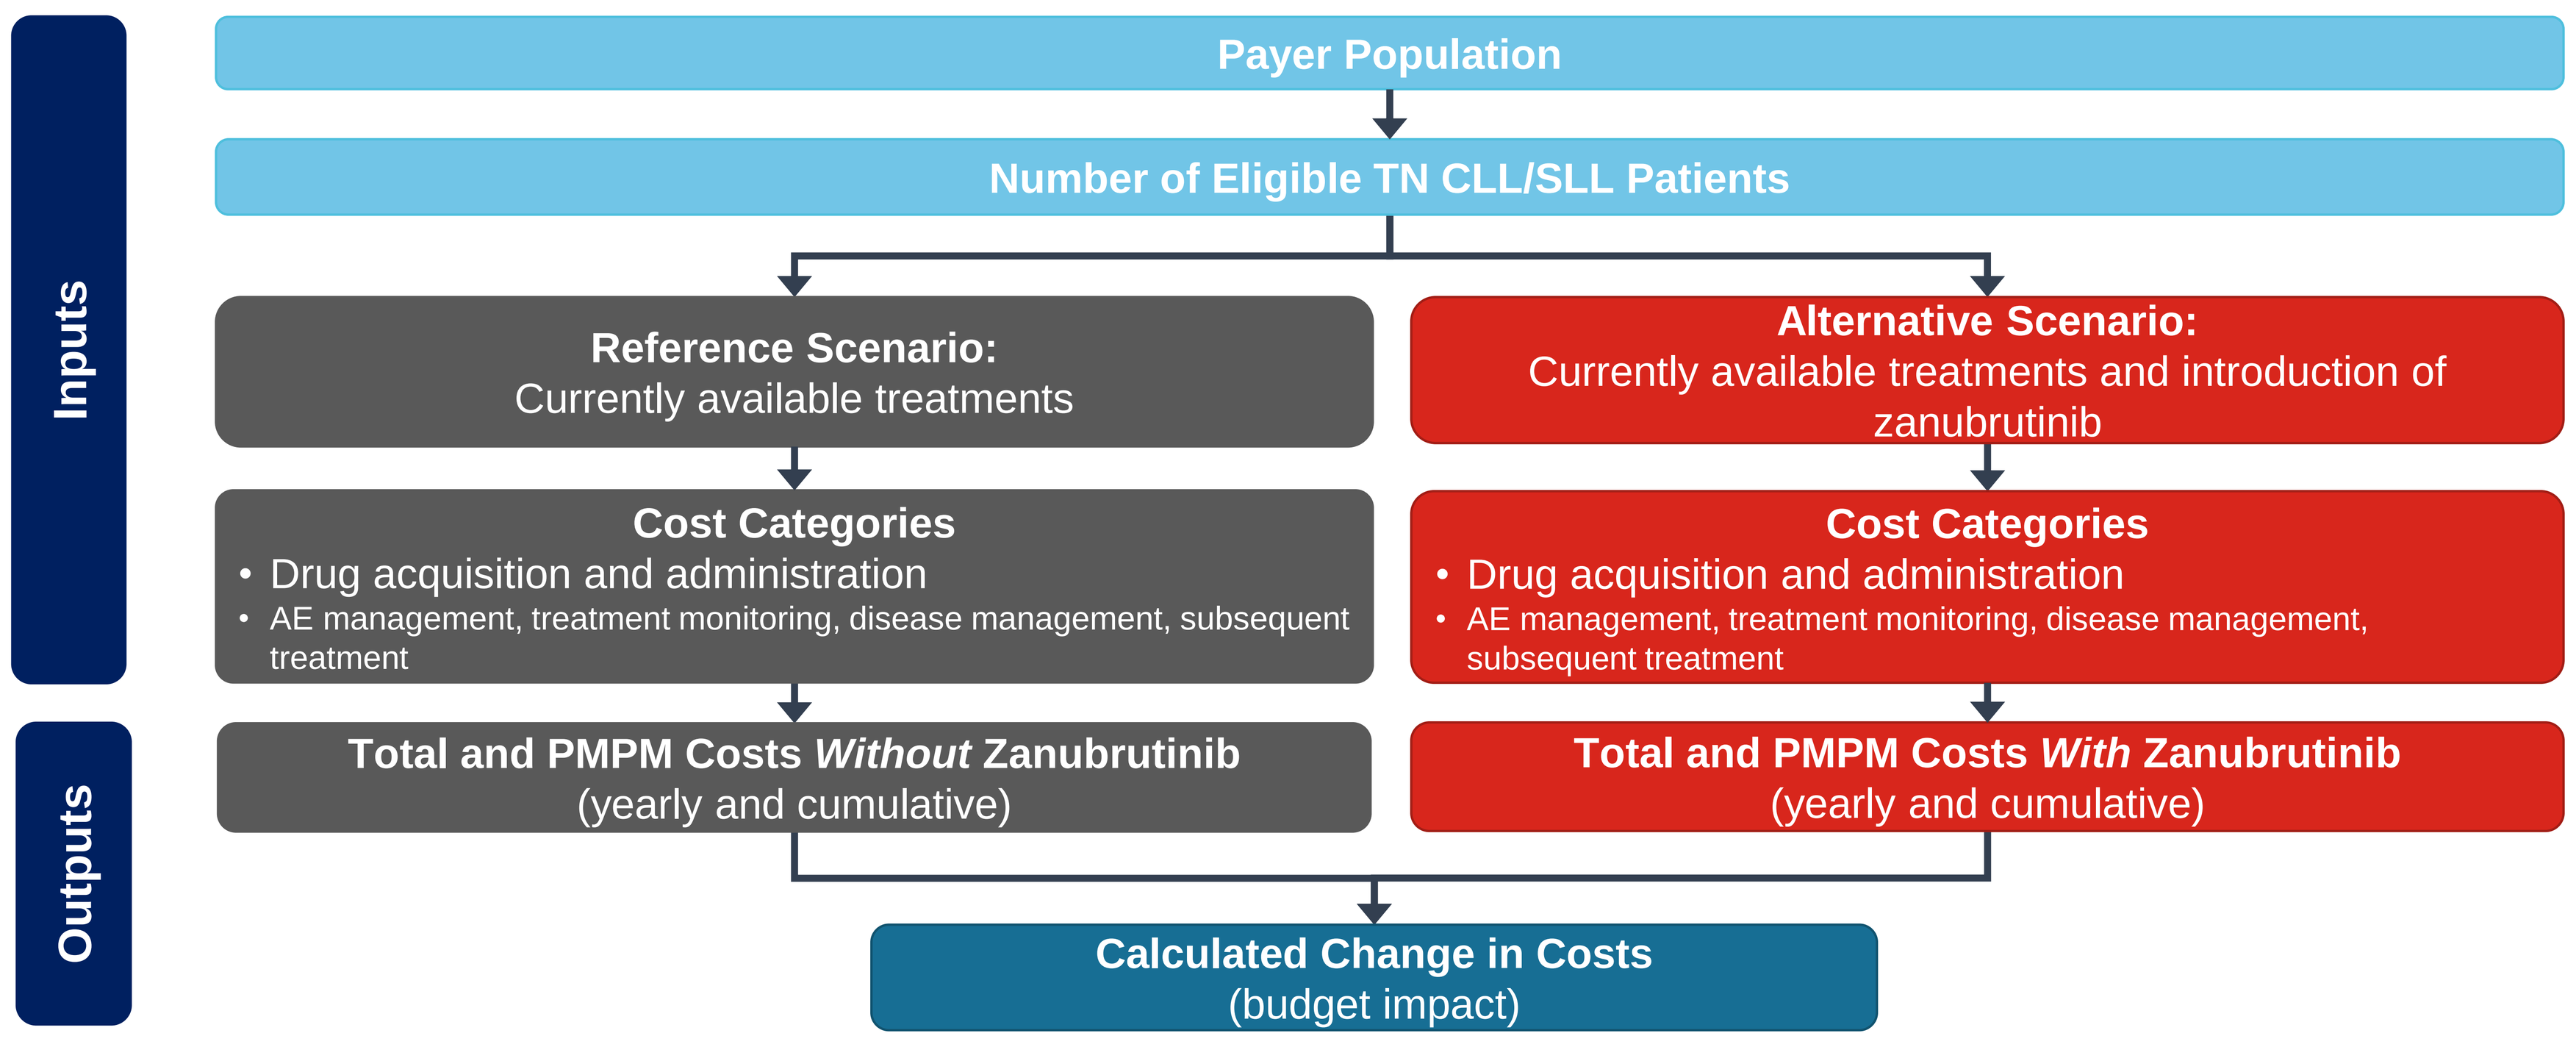
Inputs
Outputs
Payer Population
Number of Eligible TN CLL/SLL Patients
Reference Scenario:
Currently available treatments
Alternative Scenario:
Currently available treatments and introduction of zanubrutinib
Cost Categories
• Drug acquisition and administration
• AE management, treatment monitoring, disease management, subsequent treatment
Cost Categories
• Drug acquisition and administration
• AE management, treatment monitoring, disease management, subsequent treatment
Total and PMPM Costs Without Zanubrutinib
(yearly and cumulative)
Total and PMPM Costs With Zanubrutinib
(yearly and cumulative)
Calculated Change in Costs
(budget impact)

CLL/SLL = chronic lymphocytic leukemia/small lymphocytic lymphoma; PMPM = per-member-per-month; TN = treatment-native

- The targeted patients are in line with the SEQUOIA trial population and a hypothetical 1,000,000 total plan size was assumed. The number of eligible patients entering the model in each year was estimated based on US-specific epidemiological data.
- A blended payer perspective (i.e., 80% commercial and 20% Medicare) was used in base case analysis.
- Zanubrutinib was assumed to treat until progression and the treatment duration was informed by the time-to-progression (TTP) extrapolated from the observed data from SEQUOIA cohort 1 data. Treatment duration of other regimens were estimated based on the constant hazard ratios derived from a standard NMA analysis or modelled as a fixed duration based on drug labels.[5]
- Progressed patients are allowed to receive active subsequent treatments, and the costs associated with 2L were applied as one-off costs in the model, i.e., venetoclax + rituximab (V+R) was assumed for patients who progressed following BTKi whilst ibrutinib + rituximab (I+R) was assumed for patients who progressed following CITs or other combination regimens. Upon treatment discontinuation, a 4-week treatment-free interval was modelled before patients continue to the next line of treatment, to reflect clinical practice more closely.
- Budget impact and per-member-per-month (PMPM) costs were estimated for the base-case scenarios of clinical practice with and without zanubrutinib. Deterministic sensitivity analyses (DSA) were conducted by varying each input by ±20% to assess parameter uncertainties and explore key model drivers.

Model Inputs and Assumptions

- The number of eligible patients entering the model in each year was derived from a hypothetical plan size of 1 million, annual incidence by perspective, and proportion of patients eligible and receiving active 1L treatment (**Table 1**).

Table 1. Epidemiology Parameters Used in the BIM

Parameter	Value	Source
Total plan size (N)	1,000,000	Assumption
Annual incidence rate of TN CLL/SLL (per 100,000)—Commercial	4.91	SEER 21 areas (all age)[6]
Annual incidence rate of TN CLL/SLL (per 100,000)—Medicare	27.50	SEER 21 areas (age 65 and over)[6]
Proportion of TN CLL/SLL eligible for active treatment	100.0%	Assumption
Of which, percent receiving active 1L treatment (e.g., symptomatic)	32.6%	Calculated from Mato et al. 2018 (sample size of 3,214 and 1,047 received 1L)[7]

CLL/SLL = Chronic lymphocytic leukemia/small lymphocytic lymphoma; TN = treatment-native;

- Modelled treatment options for TN CLL were zanubrutinib, ibrutinib, acalabrutinib, obinutuzumab + chlorambucil, rituximab, venetoclax + obinutuzumab, bendamustine + rituximab, chlorambucil ± rituximab (**Table 2**).
- Market share data for the reference scenario were sourced from BeiGene market research.[8] Zanubrutinib uptake was assumed to be 10% and distributed proportionally to the current treatment mix. The market share values in both scenarios were assumed to be constant throughout the time horizon.
- Drug acquisition costs (commercial) were extracted from wholesale acquisition costs (WAC) reported in REDBOOK.[9] Average selling prices (ASP) published on CMS.gov were used to inform Medicare perspective. Drug wastage was considered for treatments that are dependent on weight or body surface area (BSA), as well as with relative dose intensity adjusted.
- Oral administration cost was assumed to be $0. Chemotherapy administration costs were informed by 2021 Physicians' Fee & Coding Guide (inflated to 2022).[10]
- Medical resource-use (MRU) costs include those incurred by disease management, treatment monitoring (i.e., one-off prophylaxis treatment of tumour lysis syndrome [TLS] for venetoclax-based regimens). MRU costs per month (e.g., hospitalization, emergency department visit, physician's office visit, and lab tests) were extracted from Kabadi 2020, a retrospective database analysis for patients without AEs, and inflated to 2022.[11] TLS one-off costs were calculated based on risk category as reported in MURANO trial.[12]

Methods (Cont'd)

- Adverse event (AE) management costs were applied at the start of each treatment and only grade 3+ AEs that were reported in at least 5% of patients for at least one treatment were included. AE incidences were extracted from trial publications, and costs per AE were derived from CMS.gov for Medicare and multiplied by a ratio of 2.05 for commercial.[13]

Table 2. Treatment Regimens and Cost Inputs

Category	Model Inputs
Treatment regimens included	**BTKi:** zanubrutinib (uptake assumed proportionally taken from all other existing treatment options), acalabrutinib, and ibrutinib **Venetoclax-based therapy:** venetoclax + obinutuzumab combination **Chemotherapy ± anti-CD20 therapy:** obinutuzumab ± other (i.e., obinutuzumab + chlorambucil), rituximab, bendamustine ± other (i.e., bendamustine + rituximab), and chlorambucil ± other (i.e., chlorambucil and chlorambucil + rituximab assumed),

Treatment	Drug acquisition cost (per cycle)			Drug administration cost (per cycle)			AE cost (aggregated)
(cycle = 28-day)	Cycle 1	Cycle 2-7	Cycle 8+	Cycle 1	Cycle 2-7	Cycle 8+	One-off cost
Zanubrutinib	$12,803	$12,803	$12,803	$0	$0	$0	$3,455
Ibrutinib	$14,657	$14,657	$14,657	$0	$0	$0	$6,972
Acalabrutinib	$12,897	$12,897	$12,897	$0	$0	$0	$4,390
Obinutuzumab ± Other	$20,605	$6,259	$0	$862	$211	$0	$12,553
Rituximab mono	$5,706	$3,742	$0	$440	$211	$0	$7,521
Venetoclax ± Other	$23,710	$17,790	$11,973	$862	$211	$0	$16,568
Bendamustine ± Other	$15,352	$14,232	$0	$440	$211	$0	$13,873
Chlorambucil ± Other	$3,880	$3,974	$0	$220	$106	$0	$7,521

BTKi = Bruton's Tyrosine Kinase Inhibitor; PI3Ki = phosphatidylinositol 3 kinase inhibitor;

Results

Base Case Analysis

- In a hypothetical health plan with 1,000,000 members, 31 patients were estimated to receive active treatment each year for TN CLL/SLL.
- Over a three-year time-horizon, the overall budget impact was a reduction of $82,437, representing a 0.22% cost-saving with the use of zanubrutinib (**Figure 2**). Total healthcare costs were $37.75m with zanubrutinib and $37.83m without.
- The expected average PMPM budget reduction was $0.002.

Figure 2. Overall Budget Impact Per Year

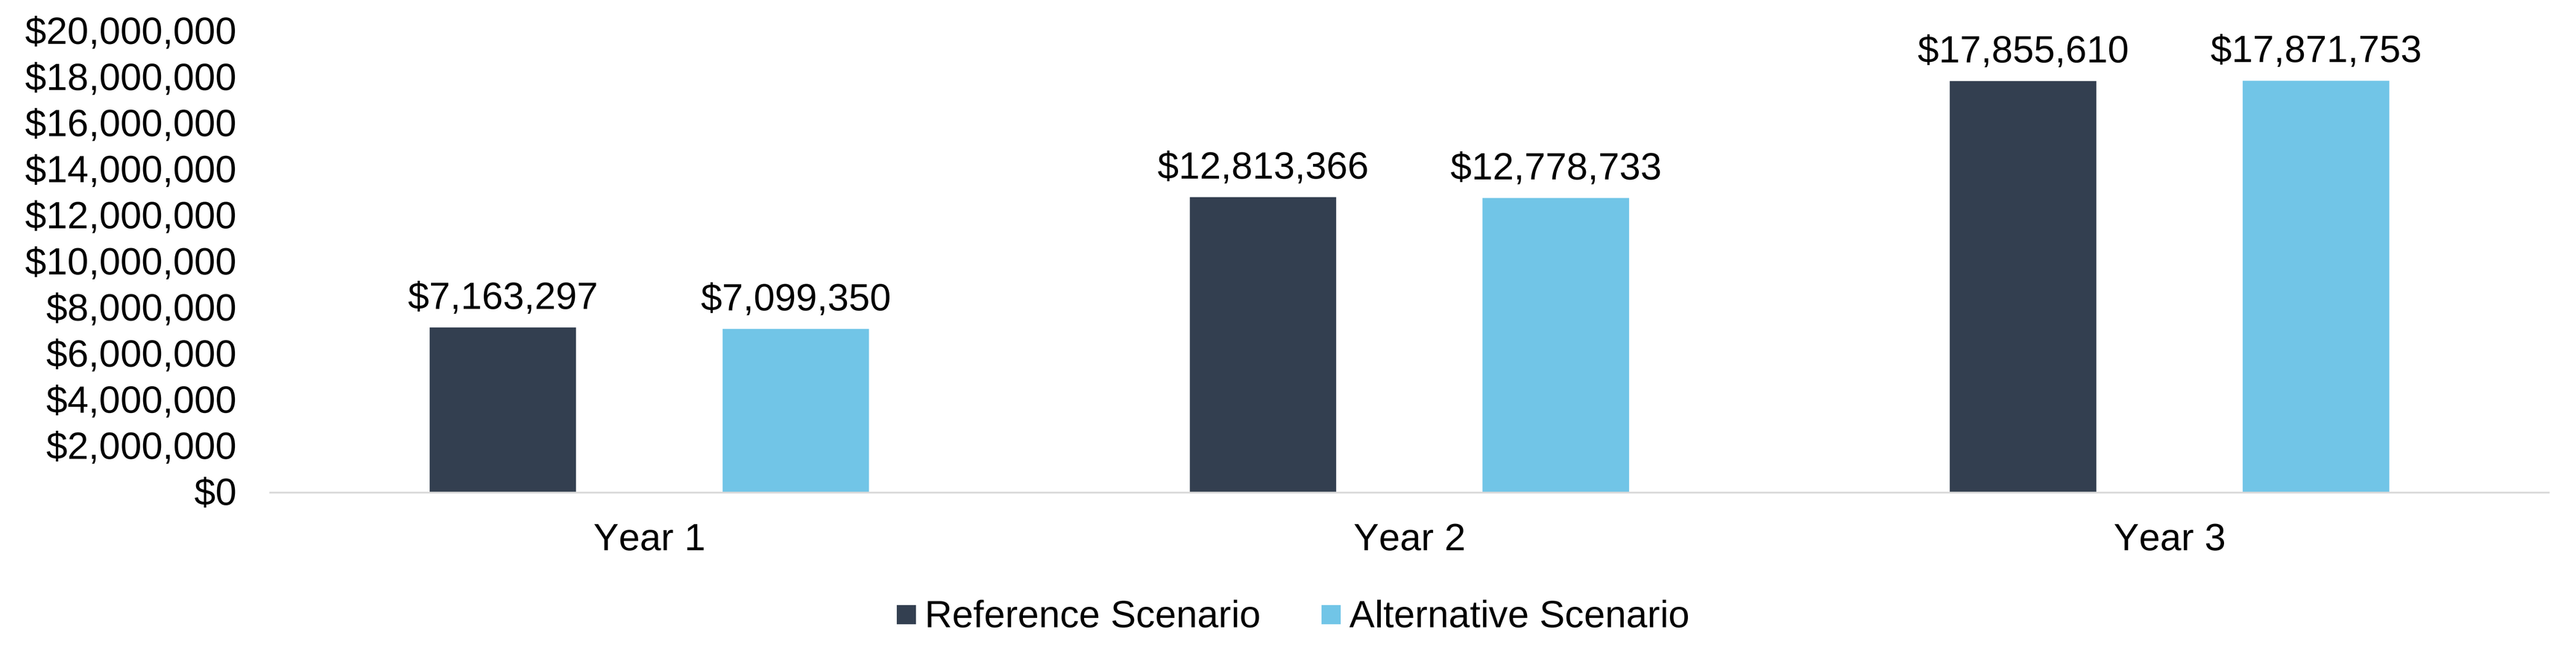
$20,000,000
$18,000,000
$16,000,000
$14,000,000
$12,000,000
$10,000,000
$8,000,000
$6,000,000
$4,000,000
$2,000,000
$0
$7,163,297
$7,099,350
$12,813,366
$12,778,733
$17,855,610
$17,871,753
Year 1
Year 2
Year 3
Reference Scenario
Alternative Scenario

Deterministic-sensitivity Analysis (DSA)

- DSA indicated that drug costs, payer perspective and treatment duration had the greatest impact on the financial budget of healthcare costs estimated over a three-year time horizon (**Figure 3**).

Figure 3. DSA Tornado Chart (Top 15 Key Drivers)

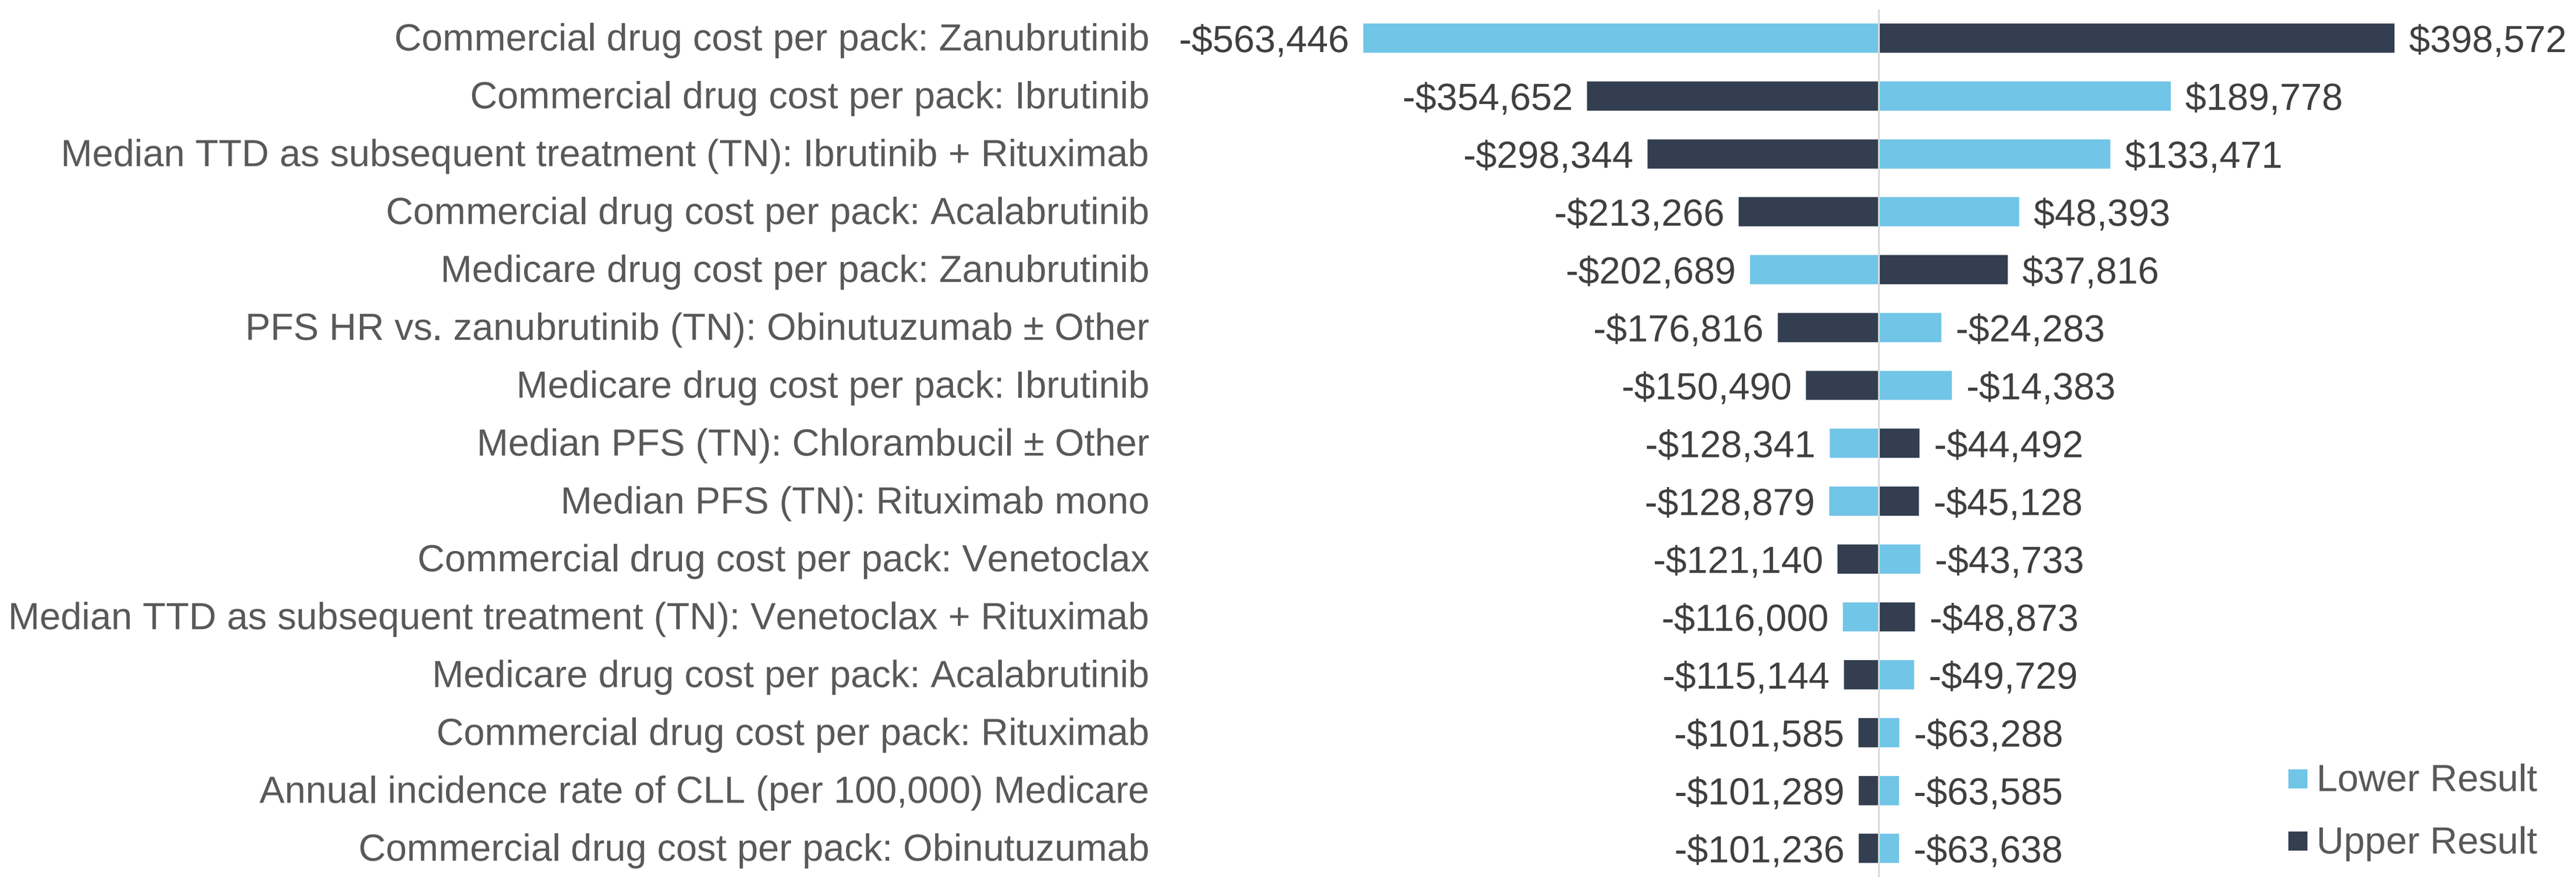
Commercial drug cost per pack: Zanubrutinib -$563,446 $398,572
Commercial drug cost per pack: Ibrutinib -$354,652 $189,778
Median TTD as subsequent treatment (TN): Ibrutinib + Rituximab -$298,344 $133,471
Commercial drug cost per pack: Acalabrutinib -$213,266 $48,393
Medicare drug cost per pack: Zanubrutinib -$202,689 $37,816
PFS HR vs. zanubrutinib (TN): Obinutuzumab ± Other -$176,816 -$24,283
Medicare drug cost per pack: Ibrutinib -$150,490 -$14,383
Median PFS (TN): Chlorambucil ± Other -$128,341 -$44,492
Median PFS (TN): Rituximab mono -$128,879 -$45,128
Commercial drug cost per pack: Venetoclax -$121,140 -$43,733
Median TTD as subsequent treatment (TN): Venetoclax + Rituximab -$116,000 -$48,873
Medicare drug cost per pack: Acalabrutinib -$115,144 -$49,729
Commercial drug cost per pack: Rituximab -$101,585 -$63,288
Annual incidence rate of CLL (per 100,000) Medicare -$101,289 -$63,585
Commercial drug cost per pack: Obinutuzumab -$101,236 -$63,638
Lower Result
Upper Result

CLL = Chronic lymphocytic leukemia; PFS = Progression-free survival; TTD = Time to treatment discontinuation; TN = Treatment-native

Limitations

- Market uptake for zanubrutinib was based on BeiGene forecasts and may be subject to future updates according to real-world utilization.
- Where treatment options could be defined as a mixed basket of monotherapy and combination therapies, changes to the default inputs impacted acquisition and administration costs but not efficacy.
- This analysis does not address any potential clinical or QoL benefits that could be associated with the inclusion of zanubrutinib in a healthcare formulary.

Conclusions

The budget impact analysis suggests that providing access to zanubrutinib for patients with TN CLL/SLL is associated with cost savings in a US health plan.

References: **1.** Herishanu Y, et al. *Hematol Oncol Clin North Am*. 2013;27(2):173-206. **2.** Hallek M. *Am J Hematol*. 2017;92(9):946-965. **3.** National Comprehensive Cancer Network (NCCN). NCCN Clinical Practice Guidelines in Oncology; Chronic Lymphocytic Leukemia/Small Lymphocytic Lymphoma; version 1.2023 NCCN,. https://www.nccn.org/professionals/physician_gls/pdf/cll.pdf. Published 2022. Updated 30 Aug 2022. Accessed 14 Sept, 2022. **4.** Clinical trial registry: https://clinicaltrials.gov/ct2/show/ NCT03336333 **5.** Chanan-Khan American Society of Clinical Oncology (ASCO); 2022 abstract (pending approval); BeiGene data on file **6.** National Cancer Institute (NCI). Registry Groupings in SEER Data and Statistics. NCI,. https://seer.cancer.gov/registries/terms.html. Published 2020. Accessed 9 Apr, 2022. **7.** Mato A, et al. *Haematologica*. 2018;103(10):e462-e465. **8.** BeiGene data on file. **9.** IBM Micromedex ® RED BOOK®. Accessed Feb 4, 2022. **10.** 2021 Physicians' Fee & Coding Guide. InHealth Professional Services. In. Atlanta, GA 30360: Commercial Fee Range. **11.** Kabadi SM, et al. *Adv Ther*. 2020;37(7):3129-3148. **12.** Seymour JF, et al. *N Engl J Med*. 2018;378(12):1107-1120. **13.** Lopez E, et al. Comparing Private Payer and Medicare Payment Rates for Select Inpatient Hospital Services. Kaiser Family Foundation (KFF). https://www.kff.org/medicare/issue-brief/comparing-private-payer-and-medicare-payment-rates-for-select-inpatient-hospital-services/. Published 2020. Updated 07 Jul 2020. Accessed 28 Mar, 2022

Presented at ISPOR EU 2022 Annual Meeting ● 6–9 November 2022 ● Vienna, Austria

Supported by funding from BeiGene USA, Inc.,