Number Needed to Treat Analysis of Zanubrutinib in Relapsed/Refractory Chronic Lymphocytic Leukemia

2337

Asher Chanan-Khan,[1] Kirollos Hanna,[2] Mei Xue,[3] Marjan Massoudi,[3] Mark Balk,[3] Ray Gani,[4] Hossein ZivariPiran,[5] Keri Yang[3]

[1]Mayo Clinic, Jacksonville, FL, USA; [2]Minnesota Oncology, St. Paul, MN, USA; [3]BeiGene USA Inc, San Mateo, CA, USA; [4]Evidera, London, UK; [5]Evidera, Toronto, Canada

BACKGROUND

- Chronic lymphocytic leukemia (CLL) is the most common leukemia type. In 2020, there were an estimated 207,463 people living with CLL, with an annual incidence of approximately 4.9 per 100,000 in the United States (US)[1]
- The goal of CLL treatment is to effectively control disease while improving/maintaining quality of life for patients, especially in patients with advanced or progressed CLL
- Following an initial response to treatment, most patients with CLL relapse and need additional therapy, while a proportion of patients become refractory to initial treatment[2]
- A recent update to NCCN clinical guidelines for the management of CLL included zanubrutinib monotherapy, ibrutinib monotherapy, acalabrutinib monotherapy, and venetoclax + rituximab as preferred treatments for relapsed/refractory (R/R) CLL without del17p/*TP53* mutation[3]
- A retrospective database analysis of commercially insured patients in the US with CLL between 2013 and 2018 found that ibrutinib was most commonly used in second-line (2L) and third-line (3L) treatment (21% and 26%, respectively)[3]
- Real-world studies indicated considerable economic burden associated with adverse events (AEs) and medical resource utilization in R/R CLL management in Medicare and commercial insurance programs[4,5]
- Zanubrutinib, a second-generation Bruton tyrosine kinase inhibitor (BTKi), demonstrated clinical superiority against ibrutinib, a first-generation BTKi, in the ALPINE trial (NCT03734016) for the treatment of adults with R/R CLL (progression-free survival [PFS] hazard ratio, 0.65; 95% CI, 0.49-0.86; *P*=0.002)[6]

OBJECTIVE

- To compare zanubrutinib versus ibrutinib in 2L R/R CLL by calculating the number needed to treat (NNT) to avoid one progression or death and associated incremental cost

METHODS

Model Overview

- An NNT analysis inside a health economic framework was conducted to characterize the number of R/R CLL patients needed to be treated with zanubrutinib instead of ibrutinib to avoid one event of progression or death. The costs for each treatment and cost differential were also calculated
- The model was developed using clinical trial data with published unit-cost and resource-use data to estimate treatment costs and PFS over a 24-month time horizon from the US payer perspective
- Payer mix was assumed with 40% commercial and 60% Medicare
- PFS was chosen as an indicator of effectiveness due to the maturity of trial data with respect to this measure and clinical interest in delaying a progression or death. Scenario analyses were conducted to test the impact of different PFS estimates and time horizons (12-month and 36-month)
- Deterministic sensitivity analyses were conducted to assess parameter uncertainties and explore key model drivers

Model Structure

- A health economic model (**Figure 1**) was used for comparing costs and outcomes for an eligible patient with R/R CLL treated with either zanubrutinib or ibrutinib as a 2L treatment

Figure 1. Structure of NNT Health Economic Model Comparing Zanubrutinib to Ibrutinib in an Eligible Patient With R/R CLL

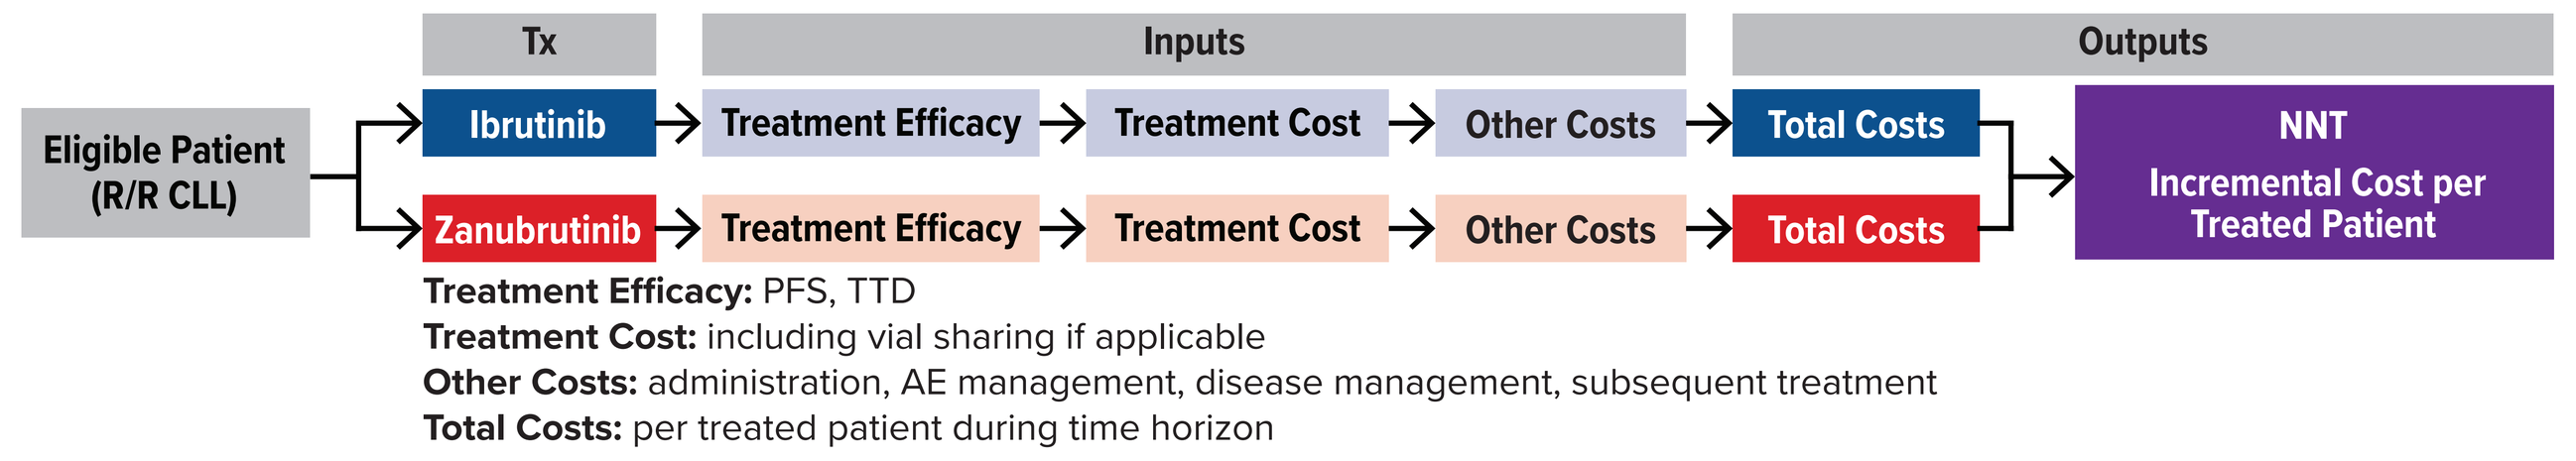
Tx
Inputs
Outputs
Eligible Patient (R/R CLL)
Ibrutinib
Treatment Efficacy
Treatment Cost
Other Costs
Total Costs
Zanubrutinib
Treatment Efficacy
Treatment Cost
Other Costs
Total Costs
NNT
Incremental Cost per Treated Patient
Treatment Efficacy: PFS, TTD
Treatment Cost: including vial sharing if applicable
Other Costs: administration, AE management, disease management, subsequent treatment
Total Costs: per treated patient during time horizon

AE, adverse event; CLL, chronic lymphocytic leukemia; NNT, number needed to treat; PFS, progression-free survival; R/R, relapsed/refractory; TTD, time to treatment discontinuation; tx, treatment.

- The model accrued patients' survival outcomes and associated costs over the time horizon for each treatment
- The 2 treatment options were compared with respect to efficacy (incremental PFS) and costs, and then NNT and cost per treated patient were calculated

MODEL INPUTS

Patient Characteristics and Clinical Inputs

- Patient characteristics and clinical inputs used in the model are shown in **Table 1**. Patient characteristics are used for calculating drug dosages and costs while the clinical inputs determine efficacy for 2L treatment with zanubrutinib and ibrutinib

Table 1. Patient Characteristics and Clinical Inputs

	Values	Sources
Patient characteristics		
Weight, kg	78.53	ALPINE study[7]
Body surface area, m^2	1.92	Calculation[8]
Clinical inputs		
PFS	Values at 12, 24, and 36 months for zanubrutinib and ibrutinib	ALPINE study[7]
TTD	Treated until progression	

PFS, progression-free survival; TTD, time to treatment discontinuation.

Treatment-Related Costs

- Costs for zanubrutinib and ibrutinib are based on both acquisition (primary 2L treatment) and administration (primary 2L treatment) costs. AE treatment costs are included in the analysis as one-off costs based on incidence in the ALPINE trial. Inpatient and outpatient resource use was based on expert opinion. Commercial and Medicare acquisition and AE costs were taken from HCUPnet and CMS.gov, respectively. Blended commercial and Medicare costs were used. All unit costs used are 2023 USD values. Values used in the model with references are shown in **Table 2**

Table 2. Treatment-Related Costs

Input	Values	Sources
Zanubrutinib	$13,521 (per 28-day cycle)	IBM Micromedex® RED BOOK® [commercial],[9] 80-mg dose[11]
Ibrutinib	$15,883 (per 28-day cycle)	IBM Micromedex® RED BOOK® [commercial],[9] 420-mg dose[12]
AE cost – zanubrutinib	$6480	ALPINE study,[7] percentages,[6] unit costs[13,14]
AE cost – ibrutinib	$6603	

AE, adverse event.

Subsequent Treatments Distribution With Costs

- Regimens and costs of subsequent treatments (3L+, acquisition, and administration) following discontinuation/stopping of 2L are included in the model. Distribution of treatment options is based on ALPINE CSR with costs blended between commercial (Redbook WAC price) and Medicare (CMS.gov). These data are shown in **Table 3**. Time on subsequent treatment was informed by median treatment duration extracted from clinical trials in R/R patients[7]

Table 3. Subsequent Treatments Distribution With Costs

Subsequent Treatment (Top 5)	Distribution (%) Zanubrutinib	Distribution (%) Ibrutinib	Cost	Sources
Percent receiving 3L+	7.3	13.8	–	ALPINE study[7]
Rituximab/rituximab-arrx	41.7	33.3	$53,083	ALPINE study,[7] cost[9,10]
Venetoclax	33.3	48.9	$349,621	ALPINE study,[7] cost[9,10]
Cyclophosphamide	20.8	13.3	$3875	ALPINE study,[7] cost[9,10]
Vincristine/vincristine sulfate	20.8	11.1	$202	ALPINE study,[7] cost[9,10]
Ibrutinib	12.5	15.6	$701,007	ALPINE study,[7] cost[9,10]

3L, third line.

Adverse Event Rates and Costs

- The model accounts for the impact of all AEs for the impact of all grade ≥3 AEs reported in ALPINE trial. All AEs were assumed to occur and be resolved in the first 4 weeks of treatment. Therefore, all AE-related costs were applied to the proportion of patients experiencing the event in the first cycle of the model. **Table 4** shows the AE event rates and costs to treat each AE

Table 4. Adverse Event Rates and Costs

Adverse Event	Incidence (%)[1,2] Zanubrutinib	Incidence (%)[1,2] Ibrutinib	Cost Commercial	Cost Medicare
Anemia	2.20	2.50	$420[15]	$420[15]
Neutropenia	21.00	18.20	$1465[15]	$1465[15]
Thrombocytopenia	3.40	5.20	$1289[15]	$1289[15]
Atrial fibrillation/flutter	2.50	4.00	$15,292[13]	$5838[14]
Hemorrhage	3.40	3.70	$19,437[13]	$9295[14]
Hypertension	15.10	13.60	$2889[16]	$2889[16]
Infection	26.50	28.10	$20,119[13]	$11,526[14]
Secondary primary malignancy	6.80	5.20	$15,043[17]	$15,043[17]

Disease-Related Healthcare Resource Use and Costs

- **Table 5** shows disease-related costs, including hospitalization, emergency department visits, office visits, laboratory and pathology, radiology, surgery, ancillary, and all other outpatient services. These are literature-based costs, based on monthly medical resource utilization[19,20] inflated to 2023[18]

Table 5. Disease-Related Healthcare Resource Use and Costs

Resource	Cost Per Treated Patient Per Month
Hospitalization	$231
Post-progression hospitalization	$1683
Emergency department visit	$15
Office visit	$137
Other services	$1214
Total: Progression free	**$1597**
Total: Post progression	**$3049**

Inputs for Scenario Analysis With Different PFS Estimates

- The key clinical parameter used in the model is PFS, which is available at 12-, 24-, and 36-month readouts from ALPINE. Time to treatment discontinuation (TTD) was capped by PFS in the model, reflecting the assumption of stopping 2L treatment upon progression. This was used for the treatment cost calculations only. For each of the PFS (12-, 24-, 36-month) estimates, an exponential curve was assumed. To investigate the sensitivity of results for different PFS estimates, data for various definitions (**Table 6**) were input into the model during the scenario analysis

Table 6. PFS at Different Time Horizons Based on Different PFS Definitions

Estimate Identifier	Zanubrutinib (% PFS) 12-month	Zanubrutinib (% PFS) 24-month	Zanubrutinib (% PFS) 36-month	Ibrutinib (% PFS) 12-month	Ibrutinib (% PFS) 24-month	Ibrutinib (% PFS) 36-month
IRC, ITT	92.5	79.5	57.9	84.8	67.3	47.2
IRC, per protocol	92.8	79.7	58.0	84.7	67.1	47.0
IRC, alternative censoring rules, ITT	92.5	80.0	58.2	84.6	67.0	**46.9**
IRC, accounting for drug interruptions, ITT	93.1	80.9	59.6	86.7	72.0	50.9
Investigator, ITT	91.3	78.4	57.5	**84.1**	65.9	49.0
Investigator, per protocol	91.5	78.6	57.7	**84.1**	**65.8**	48.9
Investigator, alternative censoring rules, ITT	**91.2**	**78.3**	**57.4**	84.3	65.9	49.0
Investigator, accounting for drug interruptions, ITT	92.5	81.2	61.3	87.7	73.1	54.2
IRC, accounting for COVID-19 death, ITT	93.4	82.0	69.2	86.2	70.0	50.3
Investigator, accounting for COVID-19 death, ITT	92.1	80.9	68.8	85.5	68.6	52.1
IRC, censored by EOT, ITT	**97.4**	**90.0**	**78.7**	**90.6**	**80.9**	**65.4**

Values in bold are maximum or minimum in each column.
EOT, end of treatment; IRC, independent review committee; ITT, intent to treat; PFS, progression-free survival.

RESULTS

Base-Case Results

- The base-case results, shown in **Table 7**, estimate that for every eight patients treated with zanubrutinib, one event of progression or death is avoided compared to using ibrutinib instead. Cost saving per patient treated with zanubrutinib is $57,330

Table 7. Base-Case Results

	24-Month PFS	Total Cost Per Treated Patient
Zanubrutinib	79.5%	$423,173
Ibrutinib	67.3%	$480,503
Incremental Results	**NNT With Zanubrutinib**	**Cost Savings With Zanubrutinib**
	8 patients	$57,330

NNT, number needed to treat; PFS, progression-free survival.

Base-Case Cost Outcomes

- The total costs per patient treated with zanubrutinib and ibrutinib are $423,173 and $480,503, respectively, with a cost saving associated with using zanubrutinib of $57,330 (**Figure 2**)
 - Drug acquisition cost is the key reason for the overall lower cost of zanubrutinib
 - Subsequent treatment cost is also lower for zanubrutinib due to improved clinical outcomes
 - Medical resource use and AE costs are comparable

Figure 2. Base-Case Disaggregated Costs for Zanubrutinib to Ibrutinib

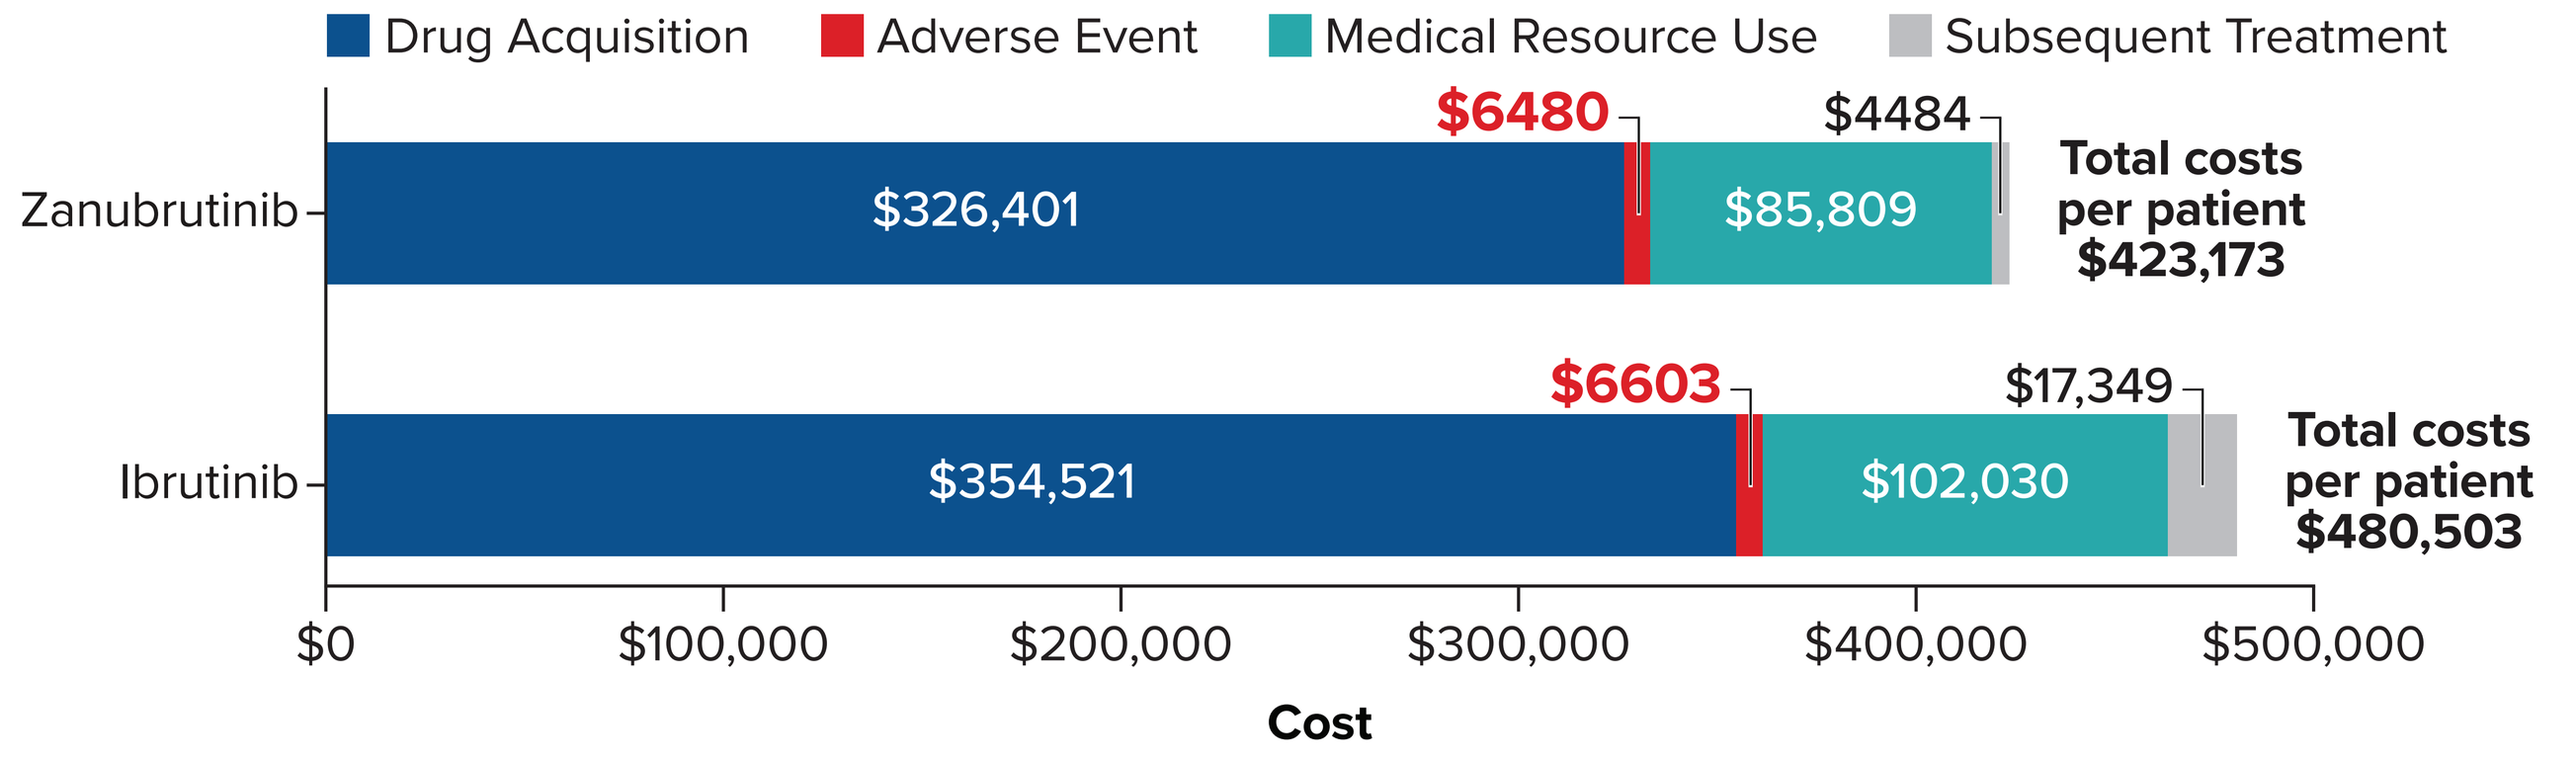
Drug Acquisition
Adverse Event
Medical Resource Use
Subsequent Treatment
Zanubrutinib
$326,401
$6480
$85,809
$4484
Total costs per patient $423,173
Ibrutinib
$354,521
$6603
$102,030
$17,349
Total costs per patient $480,503
$0
$100,000
$200,000
$300,000
$400,000
$500,000
Cost

Deterministic Sensitivity Analysis

- A deterministic sensitivity analysis (DSA) was conducted by setting the model input parameter values (one at a time) to the upper and lower bound of their reported uncertainty (95% CI or published ranges). Results are displayed in **Figure 3**
 - The DSA indicates that the model estimates are most sensitive to changes in the drug acquisition costs, with total incremental cost per patient over a 24-month period ranging from -$100,293 and -$14,368 compared to ibrutinib
 - The model is also sensitive to changes in PFS estimates for ibrutinib and zanubrutinib, while minimal impact is observed for other input changes
- Across all DSA, zanubrutinib is cost-saving

Figure 3. Deterministic Sensitivity Analysis

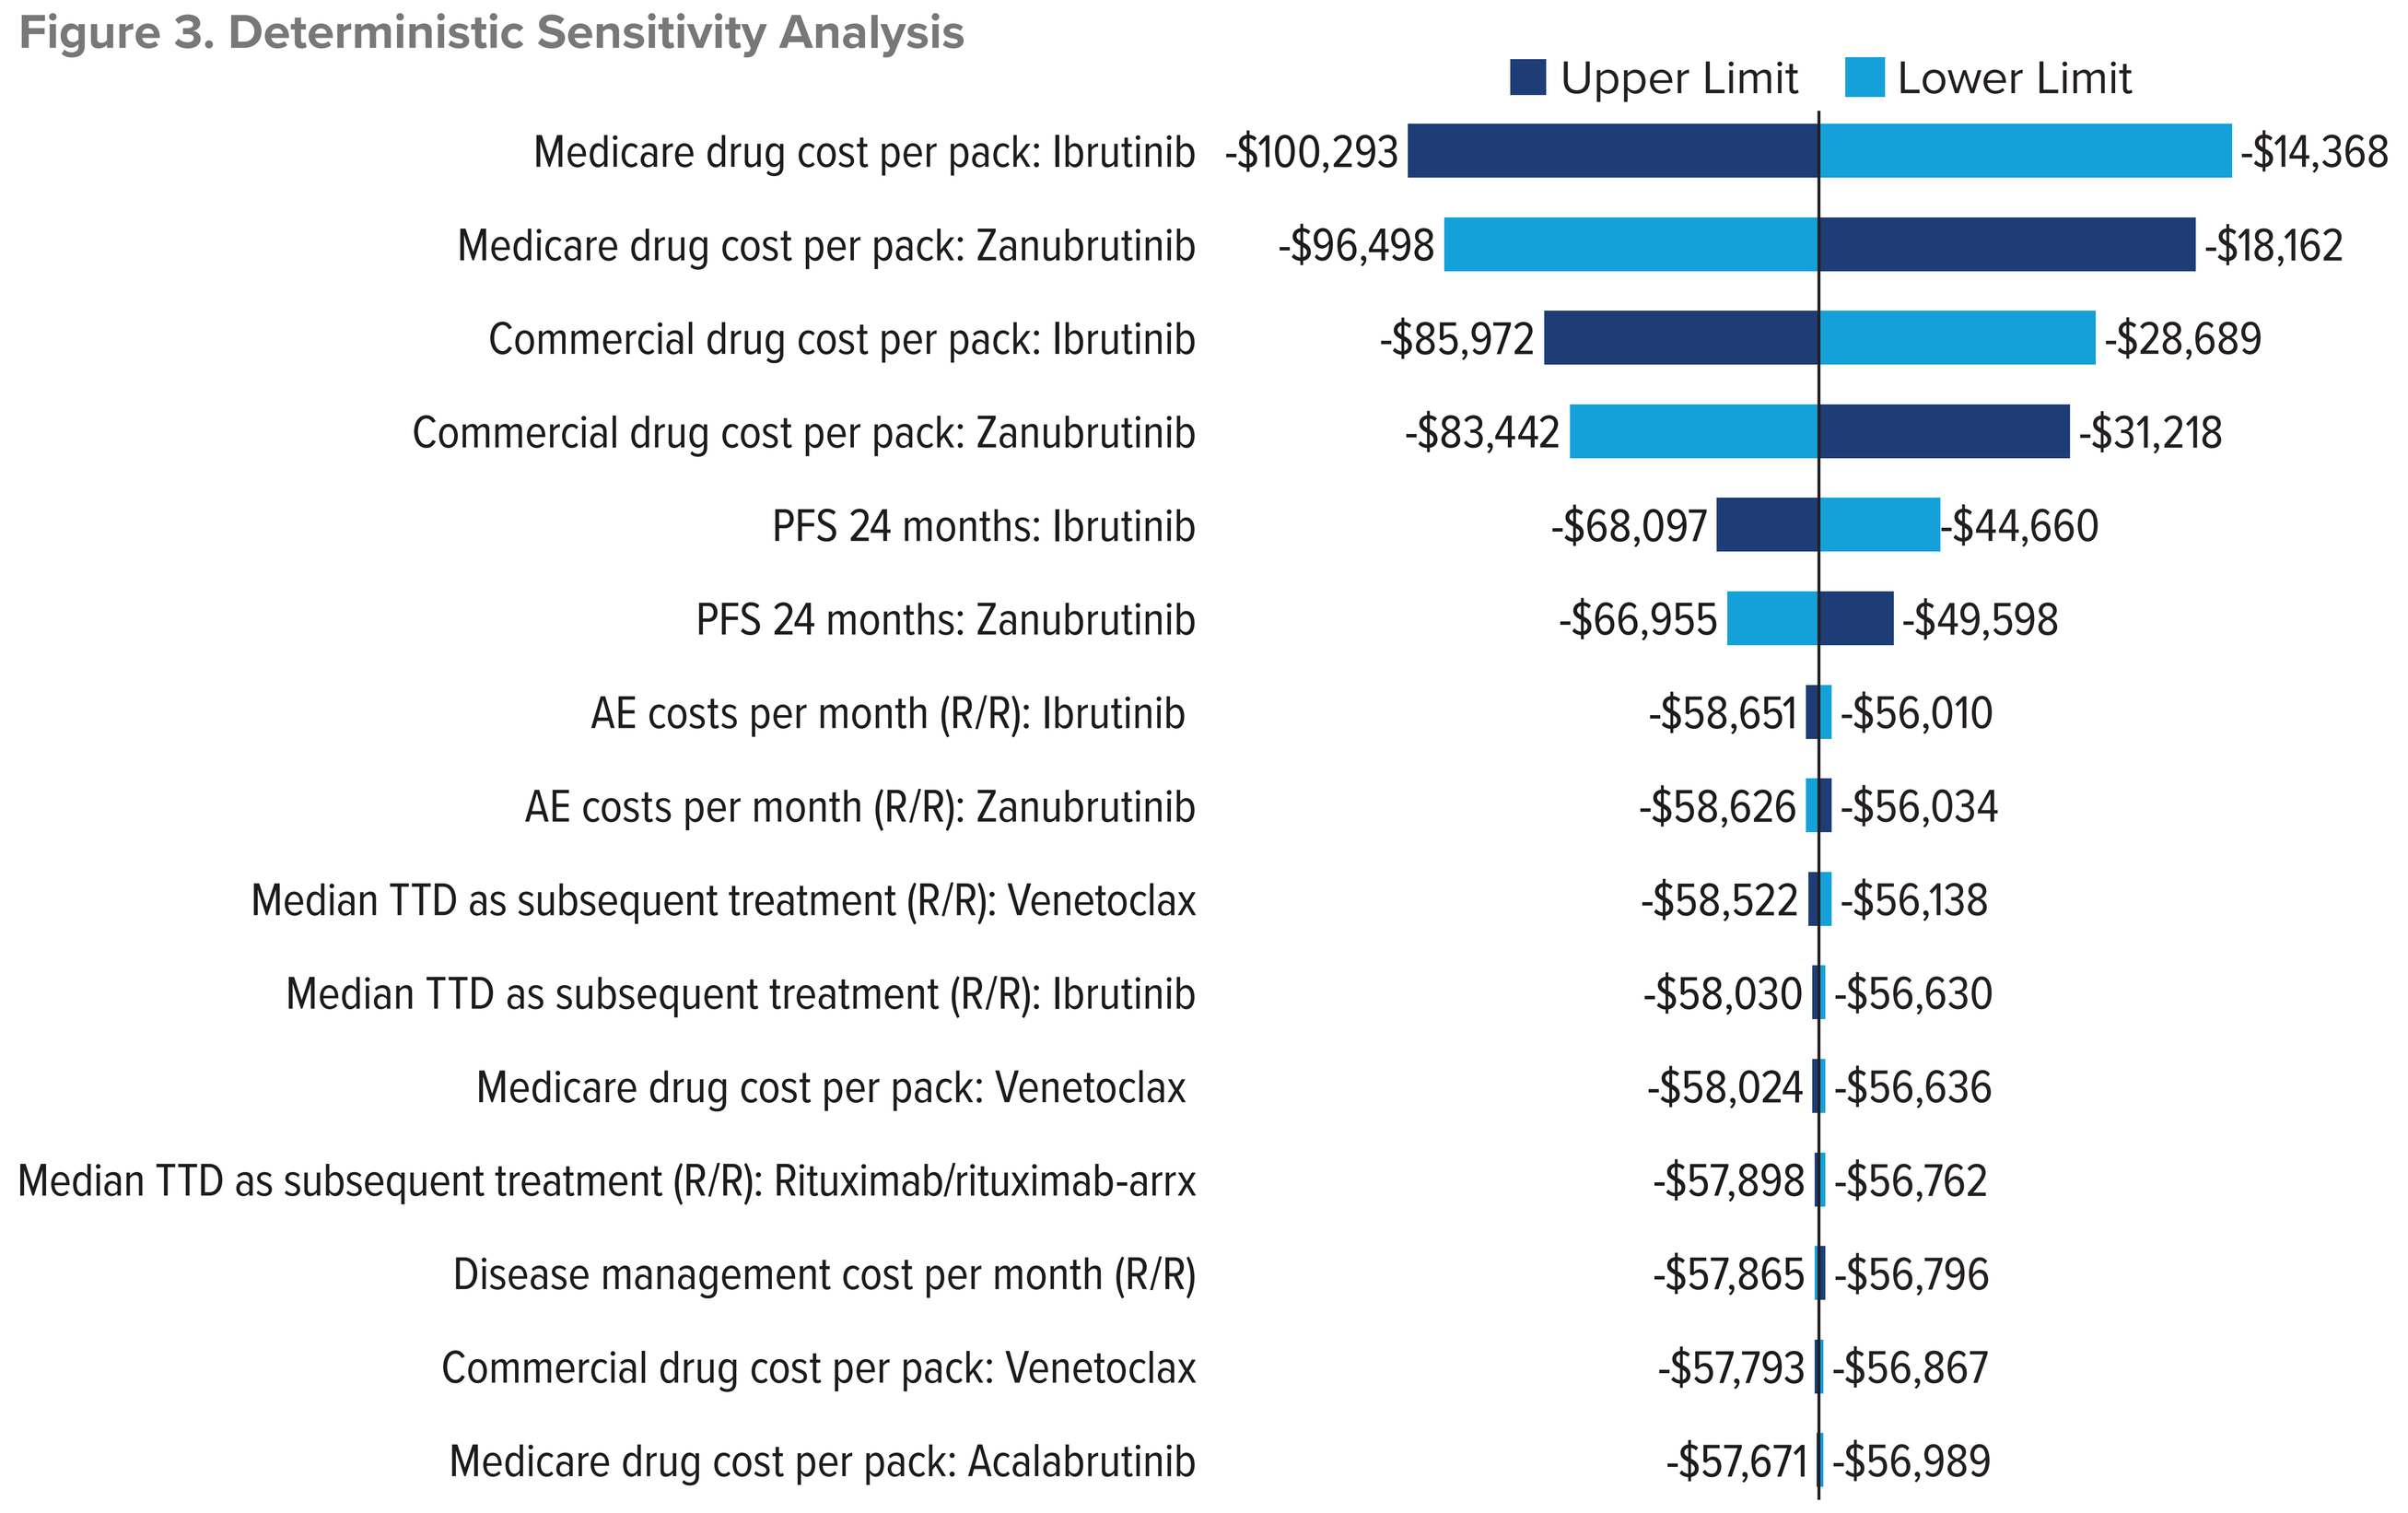
Upper Limit
Lower Limit
Medicare drug cost per pack: Ibrutinib -$100,293 -$14,368
Medicare drug cost per pack: Zanubrutinib -$96,498 -$18,162
Commercial drug cost per pack: Ibrutinib -$85,972 -$28,689
Commercial drug cost per pack: Zanubrutinib -$83,442 -$31,218
PFS 24 months: Ibrutinib -$68,097 -$44,660
PFS 24 months: Zanubrutinib -$66,955 -$49,598
AE costs per month (R/R): Ibrutinib -$58,651 -$56,010
AE costs per month (R/R): Zanubrutinib -$58,626 -$56,034
Median TTD as subsequent treatment (R/R): Venetoclax -$58,522 -$56,138
Median TTD as subsequent treatment (R/R): Ibrutinib -$58,030 -$56,630
Medicare drug cost per pack: Venetoclax -$58,024 -$56,636
Median TTD as subsequent treatment (R/R): Rituximab/rituximab-arrx -$57,898 -$56,762
Disease management cost per month (R/R) -$57,865 -$56,796
Commercial drug cost per pack: Venetoclax -$57,793 -$56,867
Medicare drug cost per pack: Acalabrutinib -$57,671 -$56,989

AE, adverse event; PFS, progression-free survival; R/R, relapsed/refractory; TTD, time to treatment discontinuation.

Scenario Analysis With Different PFS Estimates – NNT

- Scenarios were run using the PFS values in **Table 6**. The results are shown in **Figure 4**
 - All scenarios demonstrate that the NNT to prevent one event of progression or death favors zanubrutinib, ranging from 5 to 21
 - The highest NNTs are associated with drug interruptions with the lowest associated with those accounting for COVID-19 deaths
 - NNTs also tend to improve over longer time horizons as the benefits of using zanubrutinib accrue

CONCLUSIONS

- Zanubrutinib provides an alternative, second-generation BTKi option with significantly better efficacy and more favorable economic outcomes vs ibrutinib for adults with R/R CLL
- Eight patients need to be treated with zanubrutinib to avoid one event of progression or death compared with using ibrutinib
- Applying the model to a hypothetical scenario of a clinical practice of 100 patients treated with zanubrutinib vs ibrutinib suggests that approximately 13 patients will avoid disease progression events or death over 24 months and the practice would realize a savings of $5.7 million

Figure 4. Scenario Analysis – NNT Results Per PFS Scenarios and Duration

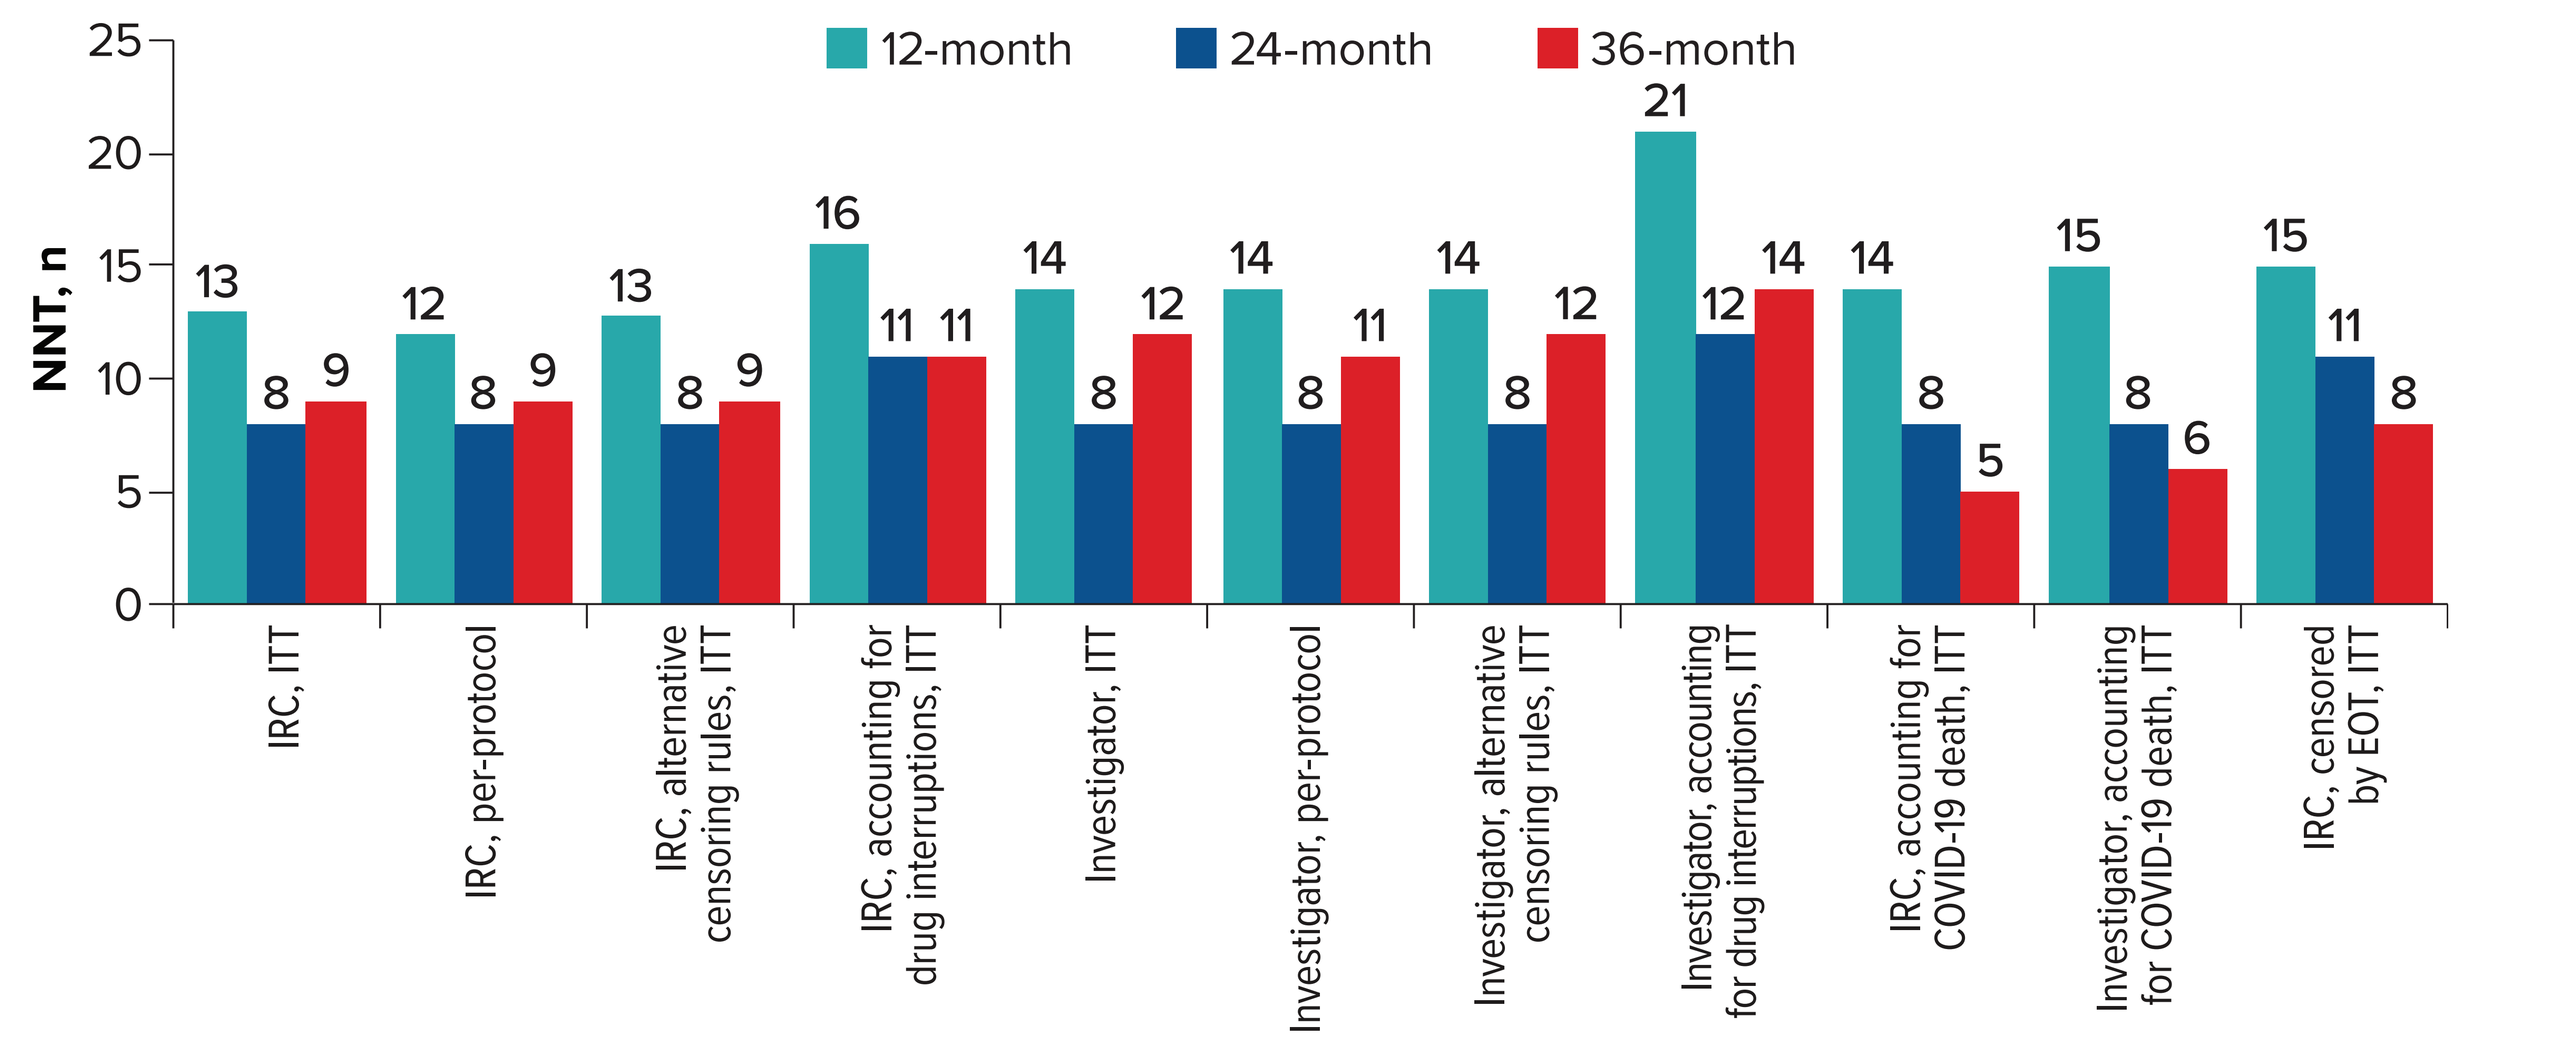
12-month
24-month
36-month
NNT, n
0
5
10
15
20
25
IRC, ITT 13 8 9
IRC, per-protocol 12 8 9
IRC, alternative censoring rules, ITT 13 8 9
IRC, accounting for drug interruptions, ITT 16 11 11
Investigator, ITT 14 8 12
Investigator, per-protocol 14 8 11
Investigator, alternative censoring rules, ITT 14 8 12
Investigator, accounting for drug interruptions, ITT 21 12 14
IRC, accounting for COVID-19 death, ITT 14 8 5
Investigator, accounting for COVID-19 death, ITT 15 8 6
IRC, censored by EOT, ITT 15 11 8

EOT, end of treatment; IRC, independent review committee; ITT, intent to treat; NNT, number needed to treat; PFS, progression-free survival.

Scenario Analysis With Different PFS Estimates – Incremental Cost Per Treated Patient

- **Figure 5** shows the cost savings associated with using the PFS values in **Table 6**
 - All scenarios demonstrate cost savings using zanubrutinib, which increase with longer time horizons
 - The greatest savings are associated with drug interruptions with the lowest associated with those accounting for COVID-19 deaths
 - The scenarios show the base-case is relatively conservative, with the alternative PFS measures leading to much greater savings

Figure 5. Scenario Analysis – Cost Per Treated Patient Per PFS Scenarios and Duration

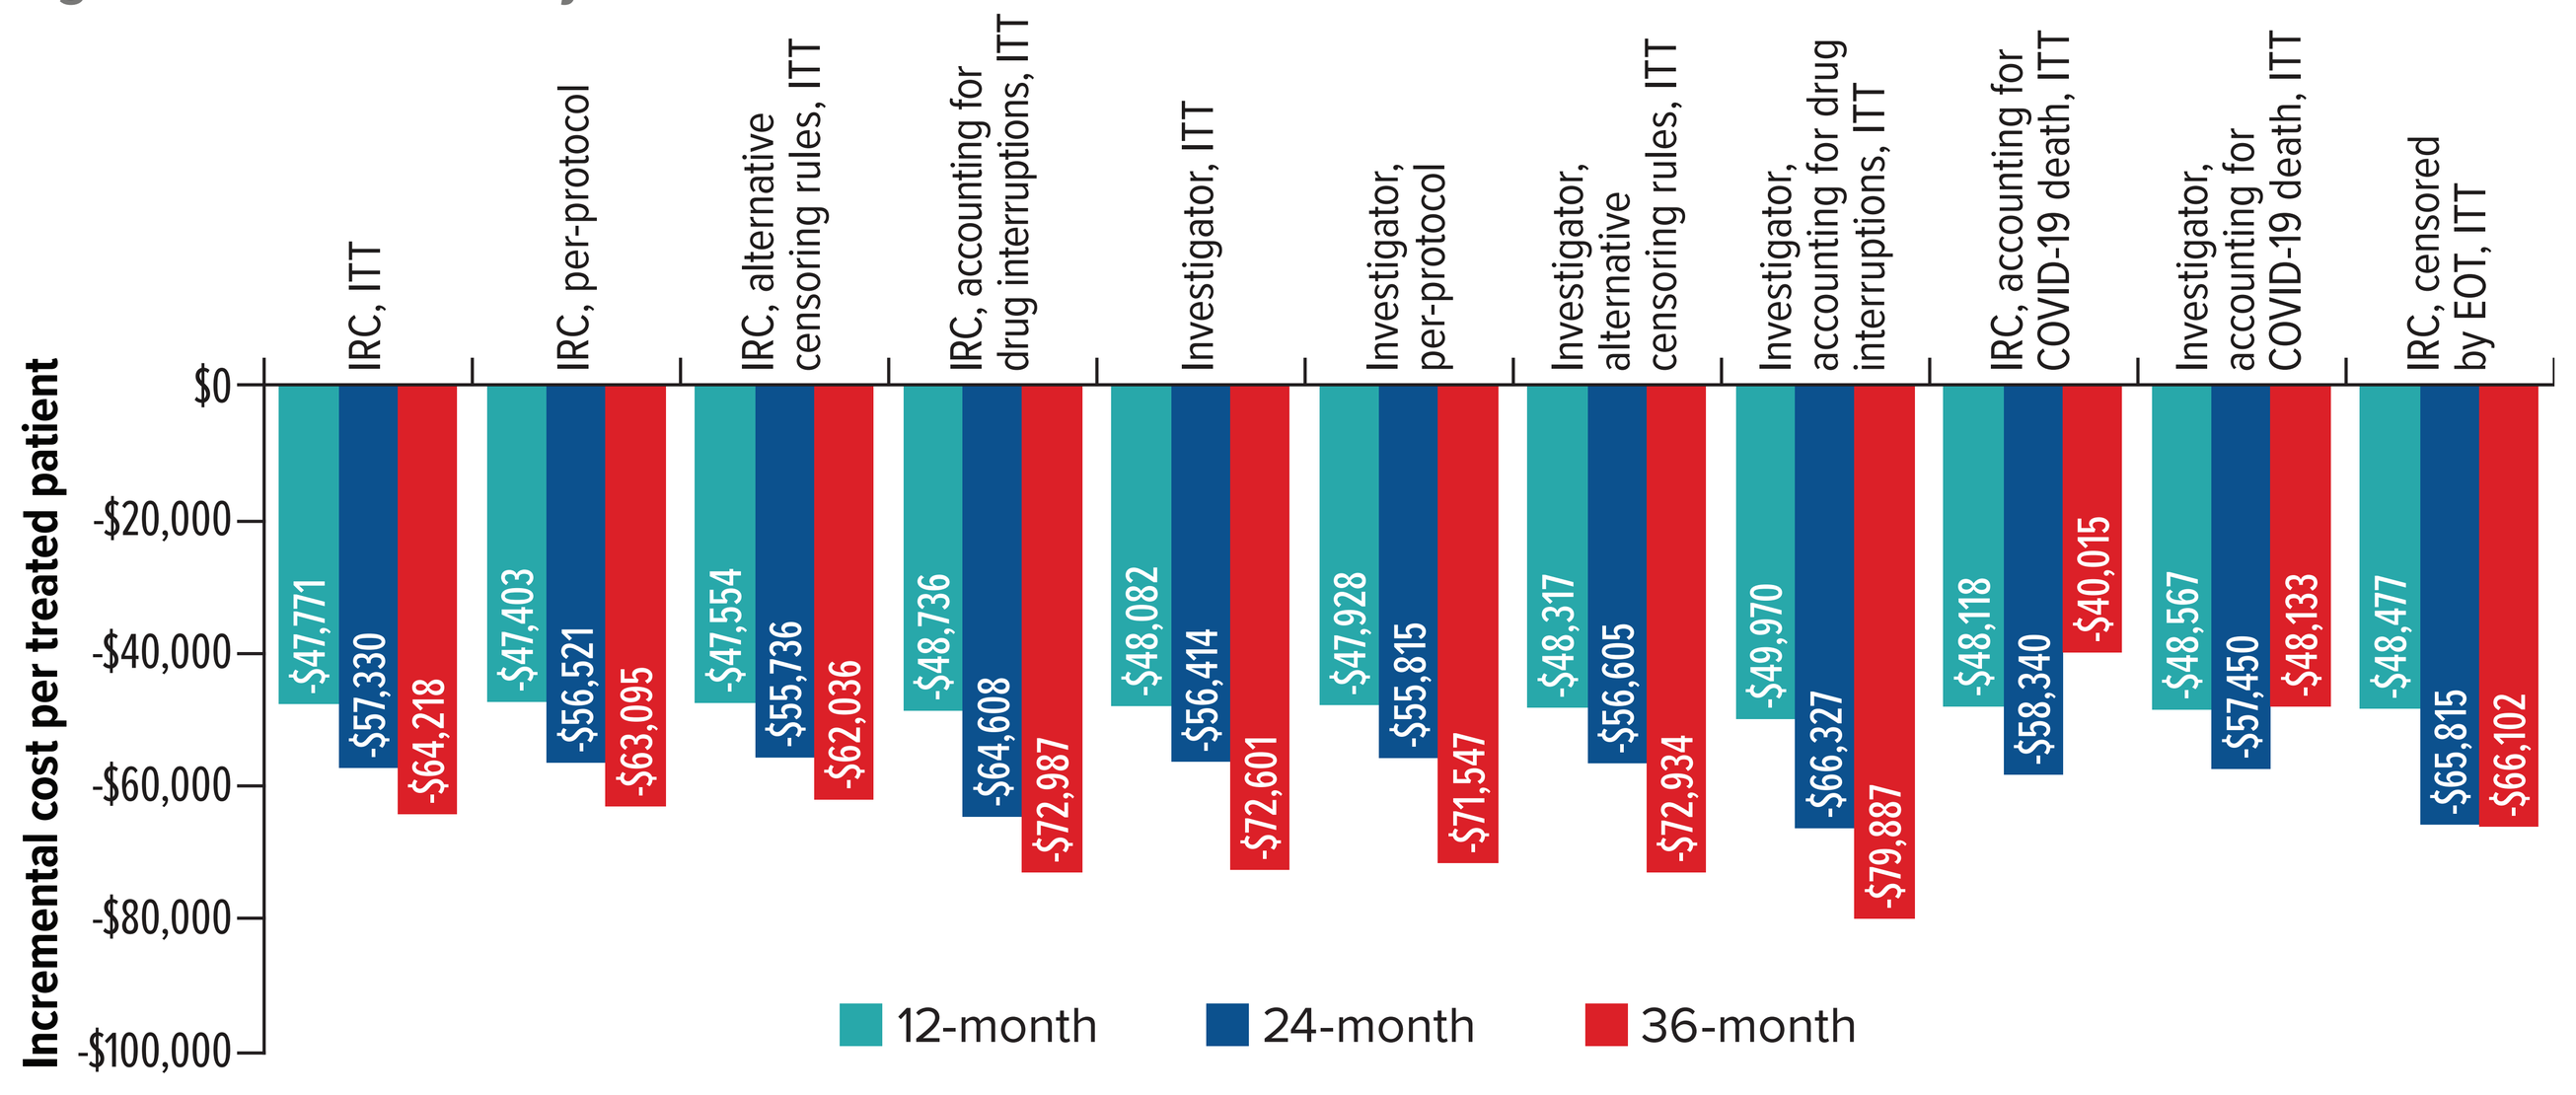
Incremental cost per treated patient
$0
-$20,000
-$40,000
-$60,000
-$80,000
-$100,000
IRC, ITT -$47,771 -$57,330 -$64,218
IRC, per-protocol -$47,403 -$56,521 -$63,095
IRC, alternative censoring rules, ITT -$47,554 -$55,736 -$62,036
IRC, accounting for drug interruptions, ITT -$48,736 -$64,608 -$72,987
Investigator, ITT -$48,082 -$56,414 -$72,601
Investigator, per-protocol -$47,928 -$55,815 -$71,547
Investigator, alternative censoring rules, ITT -$48,317 -$56,605 -$72,934
Investigator, accounting for drug interruptions, ITT -$49,970 -$66,327 -$79,887
IRC, accounting for COVID-19 death, ITT -$48,118 -$58,340 -$40,015
Investigator, accounting for COVID-19 death, ITT -$48,567 -$57,450 -$48,133
IRC, censored by EOT, ITT -$48,477 -$65,815 -$66,102
12-month
24-month
36-month

IRC, independent review committee; ITT, intent-to-treat.

REFERENCES

1. SEER Program, Registry Groupings in SEER Data and Statistics. https://seer.cancer.gov/registries/terms.html.
2. National Health Service 2019. https://www.nhs.uk/conditions/chronic-lymphocytic-leukaemia.
3. NCCN. Chronic Lymphocytic Leukemia/Small Lymphocytic Lymphoma (Version 2.2022).
4. Goyal RK, et al. *Cancer Med.* 2021;10(8):2690-2702.
5. Kabadi SM, et al. *Cancer Med.* 2019;8(8):3803-3810.
6. Brown JR, et al. *N Engl J Med.* 2023;388(4):319-332.
7. ALPINE-CSR. A phase 3, randomised study of zanubrutinib (BGB-3111) compared with ibrutinib in patients with relapsed/refractory chronic lymphocytic leukemia or small lymphocytic lymphoma.
8. Mosteller RD. *N Engl J Med.* 1987;317(17):1098.
9. IBM Micromedex® RED BOOK®.https://www.ibm.com/downloads/cas/OZKXWE1A.
10. CMS.gov. Medicare 2023 ASP Pricing File Q1 - Payment Allowance Limits for Medicare Part B Drugs. https://www.cms.gov/medicare/payment/fee-for-service-providers/part-b-drugs/average-drug-sales-price.
11. Food and Drug Administration. BRUKINSA® (zanubrutinib) capsules, for oral use. https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/213217s005lbl.pdf.
12. Food and Drug Administration. IMBRUVICA® (ibrutinib) capsules, for oral use. https://www.accessdata.fda.gov/drugsatfda_docs/label/2018/210563s000lbl.pdf.
13. AHRQ. HCUPnet Inpatient Stays, National Statistics by Clinical Classification Software Refined (CCSR), Principal Diagnosis. All Payer 2020 Cost.
14. CMS.gov. Acute Inpatient PPS. FY 2023 IPPS Final Rule Home Page. Payment by DRG Code.
15. ICER. Novel agents to prevent chemotherapy-induced neutropenia and other myelosuppressive effects. Revised evidence report. March 17, 2022.
16. Wong W, et al. *PLoS One.* 2018;13(4):e0196007.
17. van Boemmel-Wegmann S, et al. *JCO Oncol Pract.* 2022;18(1):e163-e174.
18. U.S Bureau of Labor Statistics. Medical care in U.S. city average, all urban consumers, not seasonally adjusted. 2023.
19. Kabadi SM, et al. *Adv Ther.* 2020;37(7):3129-3148.
20. Reyes C, et al. *Oncologist.* 2019;24(9):1219-1228.

ACKNOWLEDGMENTS

This study was sponsored by BeiGene, Ltd. Editorial assistance was supported by Evidera and was supported by BeiGene.

CORRESPONDENCE: Keri Yang, keri.yang@beigene.com

Presented at the 65th ASH Annual Meeting and Exposition; December 9-12, 2023; San Diego, CA